SB1317: a Potent and Orally Active FLT3-CDK inhibitor with Anti-Tumor Efficacy in Models of Acute Myeloid Leukemia

K.C. Goh, W.C. Ong, W. Stunkel, Y.C. Tan, K. Sangthongpitag, S.K. Goh, C.Y. Hu, A.L. Liang, Z. Bonday, A. William, P. Yeo, P. Venkatesh, #C.S. Chen, B. Dymock, E. Kantharaj and J.M. Wood

*S*BIO Pte Ltd, 1 Science Park Road, #05-09 The Capricorn, Singapore Science Park II, Singapore 117528*
#Yong Loo Lin School of Medicine, National University of Singapore, 10 Medical Drive, Singapore 117597

INTRODUCTION

- FLT3 is the most frequently mutated gene in acute myeloid leukemia (AML) and therefore an attractive target for novel therapies[1]
- However, limited long-term efficacy of FLT3 inhibitors in clinical development may indicate the need to target other key drivers of the disease
- Cyclin-dependent kinases 1, 2, 4 and 6 are established targets for anti-cancer drug discovery due to their direct role in cell cycle regulation and aberrations in their cyclin partners
- Transcriptional CDKs (7 and 9) are emerging as plausible anti-cancer targets due to better understanding of their effects on key apoptotic molecules
- Recent evidence suggests that the combined targeting of cell cycle and transcriptional CDKs, specifically 1, 2 and 9, confers a therapeutic advantage over individual CDK targeting[2].
- Here, we describe SB1317, a novel FLT3-CDK inhibitor that shows promising anti-tumor activity in AML models and a favorable pharmacokinetic profile for oral drug development

In Vitro Pharmacology

Table 1: SB1317 is a potent FLT3-CDK inhibitor

Family	Kinase	IC_{50} (nM)
Class III RTK	FLT3	50
	FLT3 (D835Y)	21
	Fms	27
	c-Kit, KDR, PDGFRβ	> 1000
CDK	CDK1	19
	CDK2	11
	CDK9	10

- SB1317 was tested against > 50 kinases (Upstate)
- 38 kinases were not significantly inhibited at 0.3 mM
- SB1317 demonstrates potent activity against CDKs 1, 2 and 9
- Unlike most FLT3 inhibitors in clinical trials, SB1317 shows selective inhibition within the Class III RTK family

Table 2: SB1317 shows potent anti-proliferative activity against a broad panel of tumor cells

Tumor type	Cell line	IC_{50} (nM)
AML	MV4-11	18
AML	HL60	39
B-cell lymphoma	Ramos	30
B-cell lymphoma	Karpas 1106P	61
Erythroleukemia	HEL	58
Colon	HCT116	40
Colon	Colo205	70
Prostate	PC3	43
Ovary	A2780	53
Breast	MCF7	65
Lung	H460	110
Myeloma	U266	120
Brain	U87MG	160

Cells were seeded in 96-well plates at their exponential growth phase and plates were incubated at 37°C, 5% CO_2, for 24 hr. Cells were treated in triplicates with various concentrations of SB1317 for 96 hr. Cell proliferation was monitored using the CellTiter 96® AQueous One Solution cell proliferation assay (Promega). Dose response curves were plotted using XL-fit (IDBS Ltd). The IC_{50} values are the averages of at least 2 independent experiments. CV is within ±30%.

Figure 1: SB1317 inhibits FLT3 and CDK in tumor cell lines

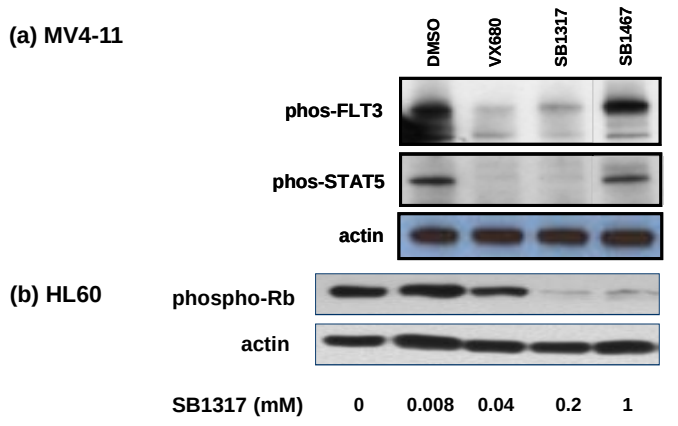
(a) MV4-11
DMSO
VX680
SB1317
SB1467
phos-FLT3
phos-STAT5
actin
(b) HL60
phospho-Rb
actin
SB1317 (mM)
0
0.008
0.04
0.2
1

(a) Sub-confluent cells were treated with vehicle or 1 mM of compounds for 4 hr. 30 mg of cell lysates were resolved on SDS-PAGE, blotted onto PVDF membranes and probed with the respective antibodies. VX-680 is a FLT3 inhibitor. SB1467 is an analog of SB1317 without FLT3 activity. (b) Sub-confluent cells were treated with various concentrations of SB1317 for 4 hr. 30 mg of cell lysates were resolved on SDS-PAGE, blotted onto PVDF membranes and probed with the respective antibodies.

Figure 2 : SB1317 induces apoptosis in tumor cell lines

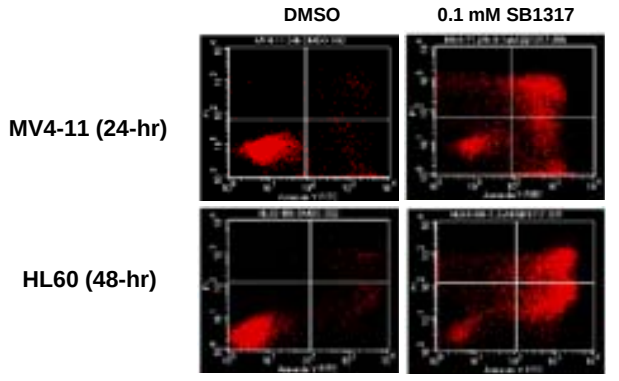
DMSO
0.1 mM SB1317
MV4-11 (24-hr)
HL60 (48-hr)

Cells were treated with the indicated concentrations of SB1317 for 24 hr. After treatment, cells were fixed, stained for DNA with propidium iodide, and analyzed on a BD Bioscience FACS Calibur cell sorter. Data were quantified using CellQuest software (BD Bioscience).

Figure 3 : SB1317 induces apoptosis in primary leukemic cells

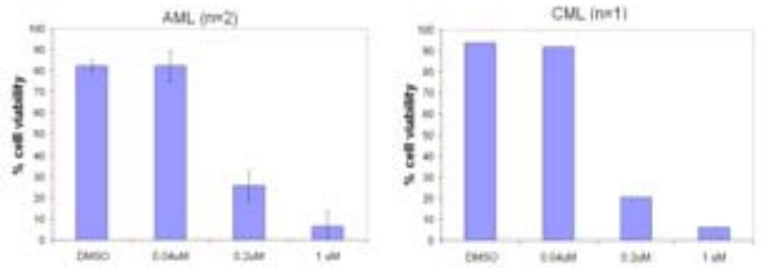
AML (n=2)
CML (n=1)
% cell viability
DMSO
0.04uM
0.2uM
1 uM

Whole blood cells from leukemia patients were plated at a density of 80,000 cells/well in a 24-well plate and treated with the indicated amounts of SB1317 for 48 hr prior to harvesting. Change in cellular viability was assessed using the Annexin V assay with FACS analysis

ADMET Profile

Figure 4 : Preclinical PK of SB1317 allows allometric scaling

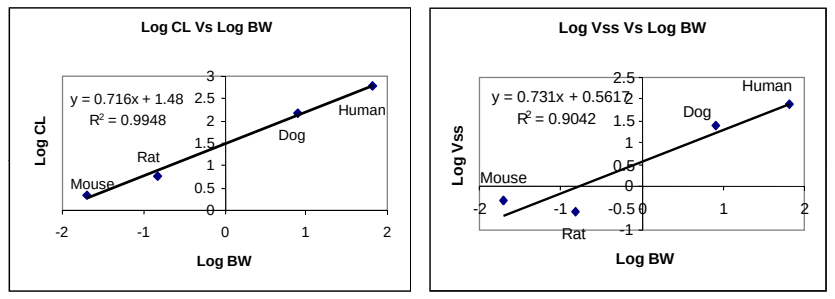
Log CL Vs Log BW
y = 0.716x + 1.48
R2 = 0.9948
Mouse
Rat
Dog
Human
Log CL
Log BW
Log Vss Vs Log BW
y = 0.731x + 0.5617
R2 = 0.9042
Log Vss

Table 3: Prediction of human dose based on preclinical PK parameters

Parameter	Mouse	Rat	Dog	Human
Body weight (kg)	0.02	0.15	8	65
V_{ss} (L)	0.46	0.27	25.1	77
CL (mL/min)	2.2	6	148	607
$t_{1/2}$ (h)	4.6	1.4	2.9	2.1
Dose (mg/kg)	5	2	1	0.5

• The predicted safe first-in-human IV dose is 30 mg. More accurate estimates for human dose can be made once NOAELs (No observed adverse effect level) is determined from toxicity studies.

Figure 5 : SB1317 exhibits dose-proportional exposure in a solid tumor model in nude mice (HCT116)

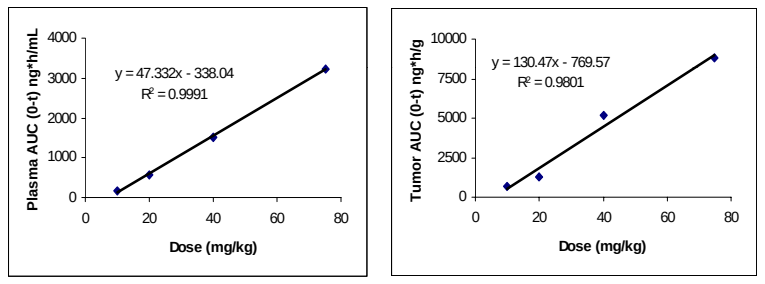
y = 47.332x - 338.04
R2 = 0.9991
Plasma AUC (0-t) ng*h/mL
Dose (mg/kg)
y = 130.47x - 769.57
R2 = 0.9801
Tumor AUC (0-t) ng*h/g

- $AUC_{(0-t)}$ tumor/plasma ratio ranges from 1.4 to 4.0 at 10, 20, 40 and 75 mg
- Dose proportional exposures in both plasma and tumor tissue

- Metabolically stable in human liver microsomes
- Highly cell permeable (Caco2 assay) and no active P-glycoprotein transport
- No P450 inhibition towards CYP3A4, 1A2, 2C9, 2C19
- No CYP3A4 induction in human hepatocytes
- Metabolized mainly by 1A2 and 3A4
- Linear pharmacokinetic extrapolation from preclinical species to human (predicted % F = 36)
- AMES test negative

In Vivo Efficacy

Figure 6 : SB1317 prevents cachexia and improves survival in an orthotopic model of AML (HL60) using oral administration

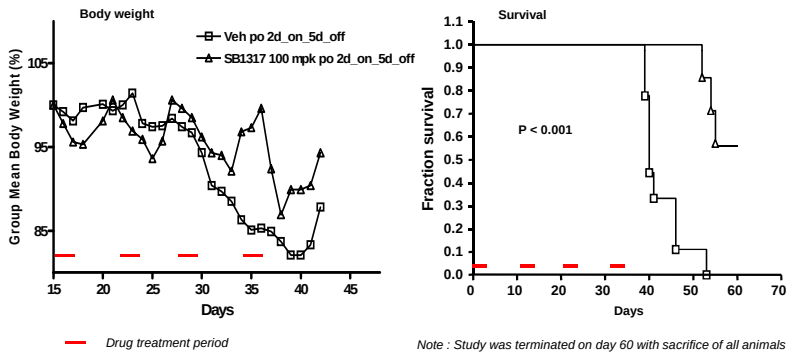
Body weight
Veh po 2d_on_5d_off
SB1317 100 mpk po 2d_on_5d_off
Group Mean Body Weight (%)
Days
Drug treatment period
Survival
Fraction survival
P < 0.001
Note : Study was terminated on day 60 with sacrifice of all animals

The HL-60 orthotopic model was established in female NOD/SCID mice by iv inoculation of 1 × 10^7 HL-60 cells at day 1. Mice were treated orally with 100 mg/kg of SB1317 or vehicle only (0.5% MC/0.1 Tween 80) starting from day 15 with a 2d_on-5d_off × 4 cycles schedule. n = 7 for the SB1317 group and n = 9 for the vehicle group. All animals were sacrificed on day 60.

Figure 7 : SB1317 prevents cachexia and improves survival in an orthotopic model of AML (HL60) using intraperitoneal administration

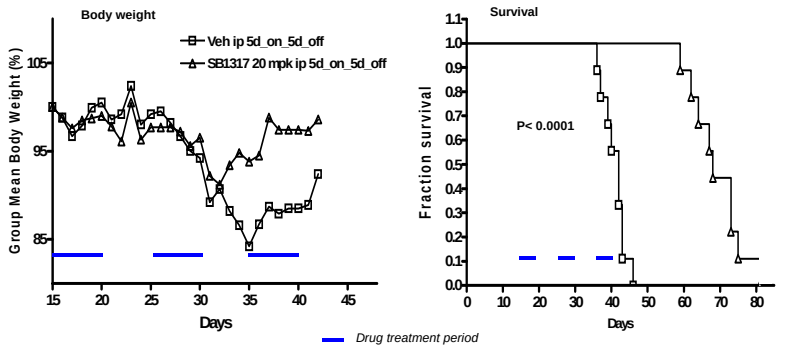
Body weight
Veh ip 5d_on_5d_off
SB1317 20 mpk ip 5d_on_5d_off
Group Mean Body Weight (%)
Days
Drug treatment period
Survival
Fraction survival
P< 0.0001

The HL-60 orthotopic model was established in female NOD/SCID mice by iv inoculation of 1 × 10^7 HL-60 cells at day 1. Mice were treated intraperitoneally with 20 mg/kg of SB1317 or vehicle only (10%DMA/10% Cremophor) starting from day 15 with a 5d_on-5d_off × 3 cycles schedule. n = 9 per group

Figure 8 : SB1317 shows excellent efficacy in a subcutaneous model of AML (MV4-11) using daily oral dosing

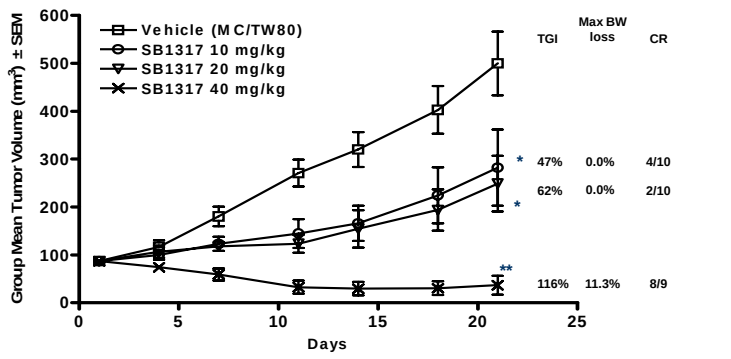
Vehicle (MC/TW80)
SB1317 10 mg/kg
SB1317 20 mg/kg
SB1317 40 mg/kg
TGI
Max BW loss
CR
47%
0.0%
4/10
62%
0.0%
2/10
116%
11.3%
8/9
Group Mean Tumor Volume (mm3) ± SEM
Days

*MV4-11 tumors were established in female BALB/c nude mice by s.c. implantation of 5 × 10^6 MV4-11 cells. All treatments were initiated on day 1 when the group mean tumor volume reached 87 mm^3 and continued until day 21. n = 9 for the 40 mg/kg SB1317 group and n=10 for the other groups. * p < 0.05, ** p < 0.01 by one-way ANOVA followed by Dunnett's Multiple Comparison Test. TGI = tumor growth inhibition, BW = body weight, CR =complete regression*

SUMMARY

SB1317 has
- A novel FLT3-CDK kinase spectrum that may be more suitable than current FLT3 compounds in development for the treatment of AML (Table 1)
- Shown potent anti-proliferative activity in a panel of tumor cell lines, especially those derived from AML (Table 2)
- Shown promising activity against primary leukemia cells (Figure 3)
- Favorable ADME properties suitable for oral dosing (Table 3 and Figure 4)
- Demonstrated excellent efficacy and tolerability in models of AML (MV4-11 and HL60, Figures 6 to 8)

CONCLUSIONS

- SB1317 exhibits potent anti-tumor activity in both *in vitro* and *in vivo* hematological tumor models
- SB1317 has favorable drug metabolism and pharmacokinetic properties.
- SB1317 shows promise as a novel oral drug for the treatment of relapsed acute leukemias and other proliferative diseases.

REFERENCES

(1) Knapper S: FLT3 inhibition in acute myeloid leukemia. *Br. J. Haem.* 2007 138:687-699

(2) Cai D, Latham VM Jr, Zhang X, Shapiro GI: Combined depletion of cell cycle and transcriptional cyclin-dependent kinase activities induces apoptosis in cancer cells. *Cancer Res.* 2006 66(18):9270-9280