SB939: a potent, orally-active HDAC inhibitor for the treatment of hematological malignancies and solid tumors

K. Sangthongpitag, H. Wang, W. Stünkel, Z. Bonday, Y. C. Tan, V. M. Nayagam, S. K. Goh, K.C. Goh, W. Xukun, X. Wu, C. Hu, E. Sun, M. Entzeroth, P. Venkatesh, E. Goh, P. Yeo, C.S. Chen, *G. Greicius, *S. Pettersson, E. Kantharaj and J. Wood

S*Bio Pte. Ltd., 1 Science Park Road #05-09 The Capricorn, Singapore Science Park II, Singapore 117528; * MTC, Division of Molecular Pathology, Laboratory of Gastrobiology, Karolinska Institute, Theorells väg 3171 77 Stockholm

INTRODUCTION

- Histone deacetylase (HDAC) regulate a number of genes involved in cell proliferation and differentiation, cell cycle progression and cell survival; important processes in tumorigenesis.
- There are a number of HDAC inhibitors (e.g. SAHA, LBH589, MGCD0103, MS275 and PXD101) emerging as a new class of anticancer agents showing efficacy for the treatment of some hematological malignancies. However clinical outcome in solid tumors has been disappointing.
- Here we present the pharmacological profiling of SB939, a novel and selective class I and class II HDAC inhibitor (Ki values ranging from 16 to 247 nM)[1&2] with improved metabolic, pharmacokinetic and pharmaceutical properties resulting in superior *in vivo* efficacy compared to other HDAC inhibitors. We present its efficacy in mouse models of early and late stage colorectal cancer as well as models of hematological malignancies.

METHODS AND RESULTS

Figure 1: Inhibition of proliferation and target inhibition in cancer cell lines

Cancer cell types	Cancer cell lines	IC_{50} (µM ± SD, n ≥ 2)
Colon	HCT116	0.91 ± 0.13
	Colo205	0.59 ± 0.11
Breast	MCF7	0.51 ± 0.10
	MDA-MB231	0.77 ± 0.17
Lung	A549	0.74± 0.10
Ovarian	A2780	0.42 ± 0.14
Prostate	PC3	0.54 ± 0.29
Leukemia	HL60	0.11 ± 0.03
	MV4-11	0.20 ± 0.06
Lymphoma	Ramos	0.14 ± 0.03
	Raji	0.15 ± 0.00

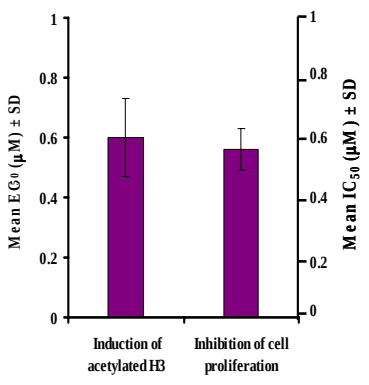
Mean EC50 (µM) ± SD
Mean IC50 (µM) ± SD
Induction of acetylated H3
Inhibition of cell proliferation

- Cell proliferation was measured using Cyquant or CellTiter 96 cell proliferation kits. Acetylation of histone3 was detected using an ELISA after 24h treatment of Colo205 cells with SB939.
- SB939 demonstrates higher potency against hematological cancer cell lines than solid tumor cell lines.
- Target inhibition (induction of acetylation of histone3, EC_{50} 0.6±0.13 µM) occurred in the same concentration range as inhibition of cell proliferation (IC_{50} 0.59 ± 0.11 µM).

Figure 2: Effects on cell cycle and cell death in (a) cancer cell lines and (b) primary cancer cells

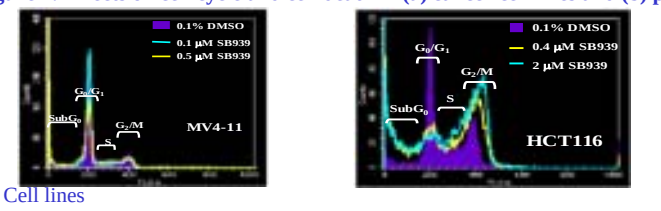
0.1% DMSO
0.1 µM SB939
0.5 µM SB939
G0/G1
SubG0
G2/M
S
MV4-11
0.1% DMSO
0.4 µM SB939
2 µM SB939
HCT116

(a) Cell lines

- After 24h treatment, HCT116 or MV4-11 cells were stained with propidium iodide and characterized using flow cytometry.
- SB939 arrested HCT116 cells at G_2/M phase, MV4-11 cells at G_1/S phase, and increased apoptotic cells at subG_0 phase.

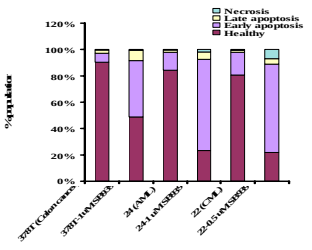
Necrosis
Late apoptosis
Early apoptosis
Healthy
%population

(b) Primary cancer

- After 48-96h treatment, the primary cancer cells were stained with Annexin V-FITC and propidium iodide and then analyzed by flow cytometry. Cell death was analyzed and quantified using cell-quest software.
- SB939 induced apoptotic cell death in primary cancer cells, including colorectal cancer and leukemia.

Figure 3: Effects on mechanistic biomarkers in HCT116 cancer cells

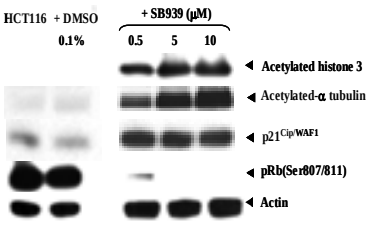
HCT116 + DMSO 0.1%
+ SB939 (µM)
0.5 5 10
Acetylated histone 3
Acetylated-α tubulin
p21Cip/WAF1
pRb(Ser807/811)
Actin

- The molecular effects of SB939 were evaluated in HCT116 cells line using western blot analysis.
- SB939 induced acetylation of histone3 and alpha tubulin, increased expression of the pro-apoptotic protein, $p21^{Cip/WAF1}$, and reduced phosphorylation of retinoblastoma protein.

Table 1: *In vitro* ADME profile

Assay	SB939
Microsomal stability ($t_{1/2}$, min)	HLM: > 60, MLM: 21 and DLM: > 60
CYP inhibition (µM)	CYP1A2, 2D6, 2C9 and 3A4: > 10 CYP2C19: 6
CYP phenotyping	Metabolized mainly by CYP3A4 and 1A2
CYP induction	No induction potential with CYP3A4 and 1A2
Solubility (mg/ml)	> 50
Caco-2	Highly permeable
	Not a P-gp substrate (ratio < 3.0)
Plasma Protein binding (% bound)	91 (human), 84 (dog), 93 (rat) and 94 (mouse)

SB939 has an excellent adsorption, distribution, metabolism and elimination (ADME) profile including:

- good metabolic stability with low potential for drug-drug interactions (no CYP inhibition or induction)
- high permeability and good solubility [Class I of biopharmaceutics classification system (BCS)].

Figure 4: Pharmacokinetic profile of SB939 in mice

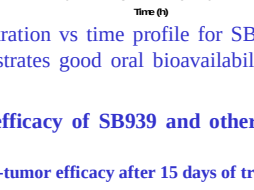
IV
PO
Plasma conc. of SB939 (ng/ml)
Time (h)

- The plasma concentration vs time profile for SB939 in mouse after 10 mg/kg (IV) and 50 mg/kg (PO) demonstrates good oral bioavailability (34%) and sufficient half life for once daily oral dosing.

Figure 5: Anti-tumor efficacy of SB939 and other HDAC inhibitors in HCT116 bearing nude mice

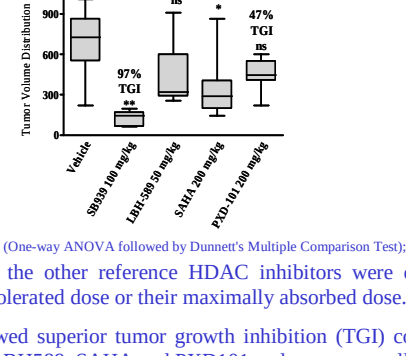
Anti-tumor efficacy after 15 days of treatment
Tumor Volume Distribution (mm3)
97% TGI **
68% TGI ns
73% TGI *
47% TGI ns
Vehicle
SB939 100 mg/kg
LBH-589 50 mg/kg
SAHA 200 mg/kg
PXD-101 200 mg/kg

*P < 0.05; **P < 0.01 (One-way ANOVA followed by Dunnett's Multiple Comparison Test); data represent medium ± range; n = 7

- SB939 and the other reference HDAC inhibitors were dosed orally once daily at their maximum tolerated dose or their maximally absorbed dose.
- SB939 showed superior tumor growth inhibition (TGI) compared to the reference HDAC inhibitors, LBH589, SAHA and PXD101 and was very well tolerated.

Figure 6: PK-PD predictors for efficacy

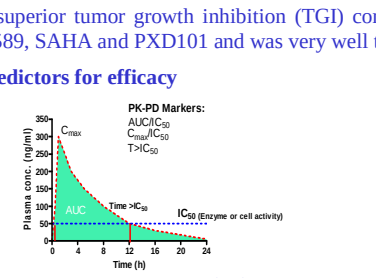
PK-PD Markers:
AUC/IC50
Cmax/IC50
T>IC50
Cmax
AUC
Time >IC50
IC50 (Enzyme or cell activity)
Plasma conc. (ng/ml)
Time (h)

→ Dependent on total dose, half-life and IC_{50} values

Figure 7: PK-PD correlation using AUC/IC_{50} (HCT116) for several different HDAC inhibitors

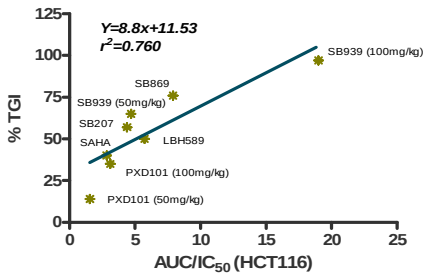
Y=8.8x+11.53
r2=0.760
SB939 (100mg/kg)
SB869
SB939 (50mg/kg)
SB207
SAHA
LBH589
PXD101 (100mg/kg)
PXD101 (50mg/kg)
% TGI
AUC/IC50 (HCT116)

- HDAC inhibitors achieving AUC/IC_{50} (HCT116) > 5 exhibited pronounced anti-tumor (PD) activity in the mouse HCT116 tumor model.
- PK-PD optimization of compounds in the preclinical setting helped us identify the best clinical candidate (SB939) and could speed up efficacy evaluation of this drug during its clinical development.

Figure 8: Target inhibition (histone3 acetylation) in HCT116 tumor tissues

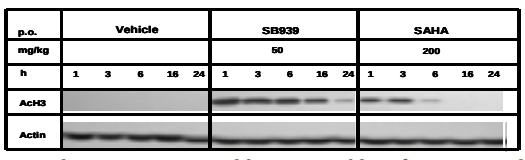
p.o.
Vehicle
SB939
SAHA
mg/kg
50
200
h
1 3 6 16 24
AcH3
Actin

- Histone acetylation was measured by western blot of tumor samples. Each blot represents tumors from 3 mice (10 µg protein each).
- After a single oral dose of SB939 (50 mg/kg) or SAHA (200 mg/kg), acetylation of histone3 was induced in tumor tissue within 1 h after dosing.
- Consistent with SB939's superior pharmacokinetic profile, the response persisted longer for SB939 than for SAHA.

Figure 9: Anti-tumor efficacy of SB939 in an early stage spontaneous colon cancer model, *Apc$^{min/+}$* Mice

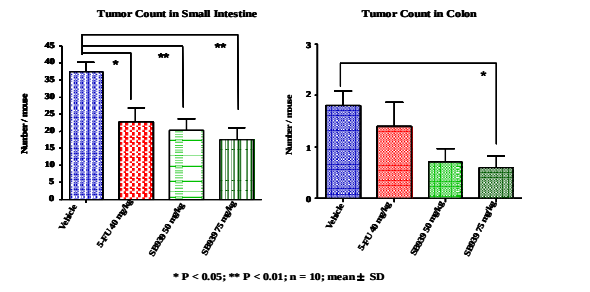
Tumor Count in Small Intestine
Tumor Count in Colon
Number / mouse
Vehicle
5-FU 40 mg/kg
SB939 50 mg/kg
SB939 75 mg/kg
* P < 0.05; ** P < 0.01; n = 10; mean ± SD

- SB939 was given orally once daily in doses of 50 or 75 mg/kg to *Apc$^{min/+}$* mice that were 16 to 20 weeks old and positive in a hemocult test, while 5FU was given by intraperitoneal administration 5 days on 10 days off for 2 cycles.
- SB939 significantly reduced tumor size and tumor number in both the small intestine and colon (46% and 61% at 50 mg/kg SB939; 53% and 67% at 75 mg/kg SB939 respectively).
- 5-FU significantly reduced tumor size and number in the small intestine by 39% but had no significant effect on lesions in the colon.

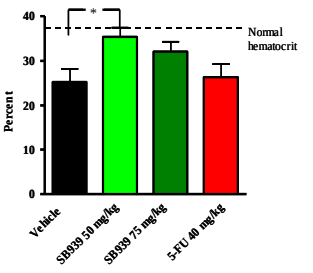
Normal hematocrit
Percent
Vehicle
SB939 50 mg/kg
SB939 75 mg/kg
5-FU 40 mg/kg

- SB939, but not 5-FU, significantly increased hematocrit and reduced the number of immature erythrocytes (Ter-119$^+$) in the spleen.

Figure 10: Anti-tumor activity of SB939 in Ramos human B-cell lymphoma tumor model

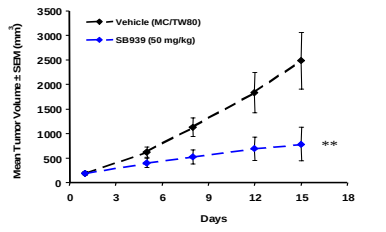
Vehicle (MC/TW80)
SB939 (50 mg/kg)
Mean Tumor Volume ± SEM (mm3)
Days

- After daily oral administration of SB939 for 14 days at 50 mg/kg, TGI was 100%. ** P < 0.01 Mann Whitney test.

Figure 11: Anti-tumor activity of SB939 in MV4-11 human acute myeloid leukemia model

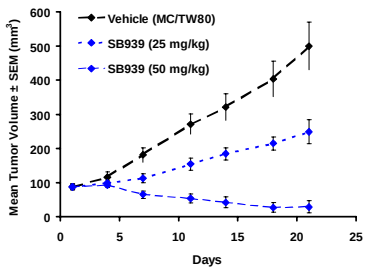
Vehicle (MC/TW80)
SB939 (25 mg/kg)
SB939 (50 mg/kg)
Mean Tumor Volume ± SEM (mm3)
Days

- After daily oral administration of SB939 for 21 days at 25 and 50 mg/kg, TGI was 59% and 116% respectively. One-way ANOVA followed by Dunnett's Multiple Comparison Test. ** P < 0.01.
- Consistent with the *in vitro* cell data, hematological tumors appear to be most sensitive to SB939.

SUMMARY

SB939

- inhibits cellular HDAC enzyme activity, resulting in induction of acetylation of histone3 and inhibition of proliferation in cancer cells and demonstrates highest potency against cells of hematological origin (Figure 1).
- arrests HCT116 at G_2/M phase and MV4-11 at G_1/S phase and induces apoptotic cell death not only in human tumor cell lines but also primary tumor cells from cancer patients (Figure 2).
- induces acetylation of histone3 and alpha tubulin and increases expression of the cyclin dependent kinase inhibitor and pro-apoptotic protein, $p21^{Cip/WAF1}$ in HCT116 colon cancer cells (Figure 3).
- is a metabolically stable, highly permeable and highly soluble compound. It does not inhibit or induce cytochrome P450 enzymes and is not a P-gp substrate (Table 1).
- has superior pharmacokinetic and pharmacodynamic properties compared to reference HDAC inhibitors (Figure 4, 6, and 7), resulting in superior anti-tumor efficacy in a model of late stage colorectal cancer (Figure 5).
- induces target inhibition (acetylation of histone3) in HCT116 tumor tissues at 1h after administration of an acute oral dose (50 mg/kg). The duration of this response persists for up to 24h and is longer than that induced by SAHA at 200 mg/kg (Figure 8).
- is also effective in a model of early stage colorectal cancer, *Apc$^{min/+}$* mice (Figure 9).
- demonstrates highest potency and efficacy in models of hematological malignancies consistent with its effects on cancer cell lines *in vitro* (Figures 10-11).

CONCLUSION

These data demonstrate that SB939 has excellent physicochemical and pharmacokinetic properties and is a potent and effective anti-tumor drug in *in vitro* and *in vivo* models of hematological malignancies and solid tumors. It has a superior profile compared to reference HDAC inhibitors that are currently in clinical trials and preclinical PK/PD modeling predicts that SB939 will have superior activity in patients. SB939 is currently being evaluated in phase I clinical trials in both hematological and solid tumors.

REFERENCES

[1]K. Sangthongpitag, H. Wang, P. Yeo, et.al. (2006) EORTC-NCI-AACR Symposium on Molecular Targets and Cancer Therapeutics Prague Congress Centre, Prague, Czech Republic, Abstract number 166

[2]K. Sangthongpitag, H. Wang, P. Yeo, et.al. (2006) EORTC-NCI-AACR Symposium on Molecular Targets and Cancer Therapeutics Prague Congress Centre, Prague, Czech Republic, Abstract number 162

ACKNOWLEDGEMENTS

We acknowledge Prof C.S. Chen, NUH, Singapore and Dr Robert Hewitt, National University of Singapore for providing us with the primary cells from the cancer patients