SB939: A potent, orally-active HDAC inhibitor for the treatment of hematologic malignancies

K. Sangthongpitag, W. Stünkel, Y.C. Tan, V.M. Nayagam, Z. Bonday, K.C. Goh, W. Xukun, S.K. Goh, X. Wu, C. Hu, H. Wang, N. Yu, E. Sun, M. Entzeroth, R. Venkatesh, P. Yeo, E. Kantharaj, C.S. Chen[1] and J. Wood

S*BIO Pte. Ltd., 1 Science Park Road #05-09 The Capricorn, Singapore Science Park II, Singapore 117528 WWW.SBIO.COM; [1]Yong Loo Lin School of Medicine, National University of Singapore, Kent Ridge Campus, Singapore, 117597

INTRODUCTION

- Histone deacetylase (HDAC) regulate a number of genes involved in cell proliferation and differentiation, cell cycle progression and cell survival; important processes in tumorigenesis.
- There are a number of HDAC inhibitors emerging as a new class of anticancer agents. Although their efficacy has been disappointing for the treatment of solid tumors, they show promising efficacy for the treatment of some hematological malignancies.
- SB939, is a novel HDAC inhibitor with superior metabolic, pharmacokinetic and pharmaceutical properties compared to other HDAC inhibitors in clinical development or currently marketed drugs such as SAHA.
- Here, we present the pharmacological profile of SB939[1,2] and demonstrate its efficacy in *in vitro* and *in vivo* models of hematological malignancies.

METHODS AND RESULTS

Table 1: Inhibition of recombinant HDAC enzymes

HDAC isoforms	K_i (nM ± SD; n ≥ 2)
HDAC1	28 ± 0.8
HDAC2	27 ± 1.4
HDAC3	19 ± 0.6
HDAC4	16 ± 0.4
HDAC5	21 ± 1.3
HDAC6	247 ± 9.9
HDAC7	104 ± 0.0
HDAC8	48 ± 1.5
HDAC9	24 ± 0.2
HDAC10	23 ± 5.6
HDAC11	43 ± 1.3
SIRT1	> 100 mM

HDAC enzymatic activity was measured using the Biomol HDAC fluorescent assay.

- SB939 potently inhibited a broad range of class I, II and IV HDACs with K_i value ranging from 16 to 247 nM.
- SB939 showed less potent inhibition of HDAC6 and HDAC7.
- SB939 did not inhibit SIRT1, a representative class III HDAC.
- SB939 did not inhibit other Zn-dependent enzymes such as MMP-3 and MMP-9.

Table 2: Inhibition of proliferation of human cancer cell lines

Cancer cell types	Cancer cell lines	IC_{50} (μM ± SD, n ≥ 2)
Colon	HCT116	0.53 ± 0.24
	Colo205	0.59 ± 0.11
Breast	MCF7	0.51 ± 0.10
	MDA-MB231	0.77 ± 0.17
Ovarian	A2780	0.42 ± 0.14
	SKOV3	2.20 ± 0.85
Prostate	PC3	0.54 ± 0.29
	DU145	0.53 ± 0.06
Liver	Hep3B	1.20 ± 0.81
	SK-Hep-1	0.70 ± 0.38
Renal	ACHN	1.20 ± 0.01
	A498	1.80 ± 0.71
Lung	A549	0.74± 0.10
	H460	1.20± 0.38
Lymphoma	**Ramos**	**0.14 ± 0.03**
	Raji	**0.15 ± 0.00**
Leukemia	**HL60**	**0.11 ± 0.03**
	MV4-11	**0.20 ± 0.06**
	IM-9	**0.09 ± 0.00**
	MOLT-4	**0.13 ± 0.06**
	K562	**0.26 ± 0.02**
	Hut78	**0.20 ± 0.07**
	MJ	**0.06 ± 0.02**
	HH	**0.09 ± 0.02**

Cell proliferation was measured using Cyquant or CellTiter96 cell proliferation kits after 96h of treatment.

- SB939 inhibits the proliferation of a broad range of tumor cell lines. Cancer cells of **hematological** origin are the most sensitive.

Figure 1: Effects on cell cycle and cell death in and acute myeloid leukemia (AML) cell line, MV4-11

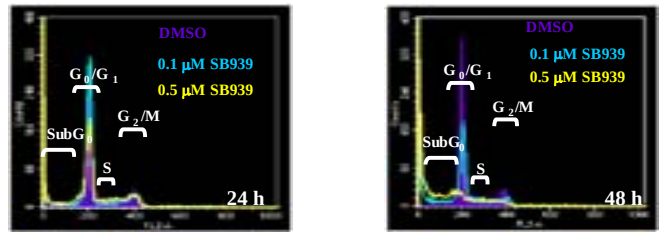
DMSO
0.1 μM SB939
0.5 μM SB939
G_0/G_1
G_2/M
$SubG_0$
S
24 h
48 h

MV4-11 AML cancer cells were treated with SB939 for 24 h and 48 h. Cells were stained with propidium iodide and analyzed by FACS.

- SB939 arrested MV4-11 cells at G_0/G_1 phase and increased cell death at $subG_0$ in dose- and time-dependent manner.

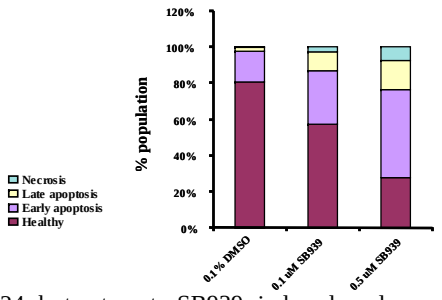
Necrosis
Late apoptosis
Early apoptosis
Healthy
% population
120%
100%
80%
60%
40%
20%
0%
0.1% DMSO
0.1 uM SB939
0.5 uM SB939

Apoptotic induction was confirmed using Annexin V-FITC and propidium iodide, and then analyzed and quantified using cell-quest software.

- After 24 h treatment, SB939 induced early apoptotic cell death in a dose-dependent manner.

Figure 2: Effects of SB939 and other HDAC inhibitors on cell viability of primary cells from an AML and a CML patient

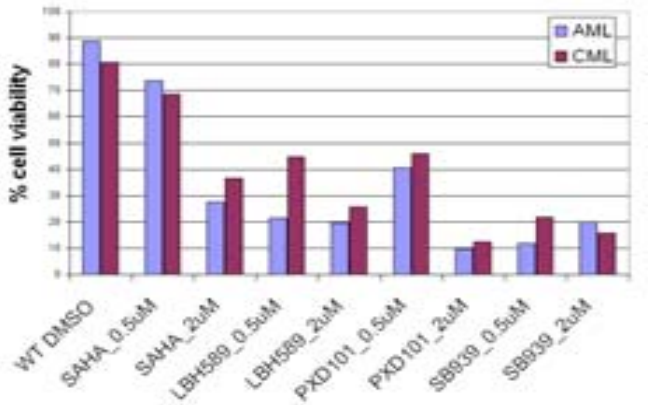
AML
CML
% cell viability
WT DMSO
SAHA_0.5uM
SAHA_2uM
LBH589_0.5uM
LBH589_2uM
PXD101_0.5uM
PXD101_2uM
SB939_0.5uM
SB939_2uM

The primary cancer cells were treated with SB939, SAHA, LBH589 and PXD101 at different concentrations. Cells were subsequently stained with Annexin V-FITC and propidium iodide and then analyzed by flow cytometry. Cell viability was analyzed and quantified using cell-quest software.

- SB939 more potently induced apoptotic cell death of primary leukemic cancer cells than SAHA, LBH589 and PXD101.

Table 3: *In vitro* ADME profile

Assay	SB939
Microsomal stability ($t_{1/2}$, min)	HLM: > 60, MLM: 21 and DLM: > 60
CYP inhibition (μM)	CYP1A2, 2D6, 2C9, 2C19, 3A4: > 5
CYP phenotyping	Metabolized mainly by CYP3A4 and 1A2
CYP induction	No induction potential with CYP3A4 and 1A2
Solubility (mg/ml)	> 50
Caco-2 (x10^{-6} cm/s)	Highly permeable
	Not a P-gp substrate (ratio < 3.0)
Plasma Protein binding (% bound)	91 (human), 84 (dog), 93 (rat) and 94 (mouse)

SB939 has an excellent adsorption, distribution, metabolism and elimination (ADME) profile including:

- good metabolic stability with low potential for drug-drug interactions (no CYP inhibition or induction)
- high permeability and good solubility [Class I of biopharmaceutics classification system (BCS)].

Pharmacokinetic profile on SB939 in mice (data not shown): the plasma concentration against time profile for SB939 in mouse after 10 mg/kg (IV) and 50 mg/kg (PO) demonstrates good oral bioavailability (34%) and sufficient half life for once daily oral dosing.

Table 4: Efficacy of SB939 in human xenograft models

Mouse Model	Tumor cells	%TGI at dose (mg/kg)		
		25	50	100
Colon	HCT116	37	61*	97**
Ovarian	A2780	-	44	68**
Prostate	PC3	-	22	46*
Lymphoma	**Ramos**	**-**	**100****	**-**
Leukemia	**MV4-11**	**59****	**116****	**-**

Female athymic nude mice, 12-14 weeks of age, were implanted subcutaneously in the flank with human cancer cells. When the tumor reached approximately 100 mm^3 in size, the mice were pair-matched into various treatment groups. The tumor-bearing mice were doses orally once daily oral with SB939 at different doses for up to 21 days. Tumors were measured 3 times a week and animals were weighted twice weekly for the duration of study. Tumor growth inhibition (TGI) was analyzed. One-way ANOVA followed by Dunnett's Multiple Comparison test (multiple groups) or Mann Whitney test (2 groups) was used to determine the statistical significance of median tumor volume between a treatment group and the vehicle control group. ** $P < 0.01$, *$P < 0.05$

- SB939 was well tolerated.
- SB939 induced dose-depended inhibition of tumor growth in HCT116, A2780, PC3 and MV4-11.
- SB939 at a dose of 50 mg/kg induced regression in the MV4-11 AML model and growth arrest in the Ramos lymphoma model
- Consistent with the *in vitro* cell data, hematological tumors appear to be most sensitive to SB939 in *in vivo* xenograft models.

Figure 3: Efficacy of SB939 in MV4-11 human acute myeloid leukemia model

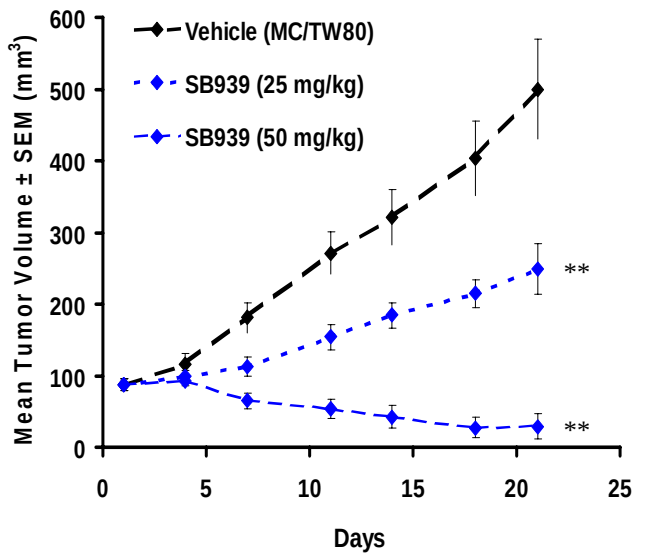
Vehicle (MC/TW80)
SB939 (25 mg/kg)
SB939 (50 mg/kg)
Mean Tumor Volume ± SEM (mm^3)
Days
**
**

Nude mice bearing MV4-11 tumors were given 25 or 50 mg/kg SB939 p.o. daily for 21 days. Data shown are mean ± SEM (n=9-10).

- SB939 induced complete tumor regression (CR) in 1 out of 10 (1/10) in the 25 mg/kg group, whereas 6/10 CR and 1/10 partial tumor regression (PR) was observed in the 50 mg/kg group.
- For all CR mice, only one mouse in the 50 mg/kg group had tumor re-growth 28 days after the last dosing.
- The tumor growth delay was 18 days (p=0.024) and 27 days (p=0.0002) at 25 and 50 mg/kg respectively.

Figure 4: Efficacy of SB939 in Ramos human B-cell lymphoma tumor model

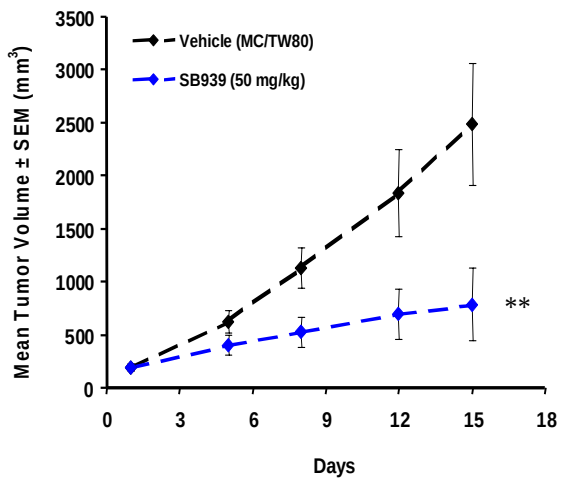
Vehicle (MC/TW80)
SB939 (50 mg/kg)
Mean Tumor Volume ± SEM (mm^3)
Days
**

Nude mice bearing Ramos tumors were given 50 mg/kg SB939 p.o. daily for 14 days. Data shown are mean ± SEM (n=10).

- During treatment, SB939 induced complete tumor regression (CR) in 3 out of 10 (3/10) mice in the 50 mg/kg group.

SUMMARY

SB939:

- potently inhibits a broad range of class I, II and IV HDACs (Table 1).
- inhibits the proliferation of a broad range of cancer cell lines and demonstrates highest potency against cells of hematological origin (Table 2).
- arrests MV4-11 cells at G_0/G_1 phase leading to apoptotic cell death in dose- and time- dependent manner (Figure 1).
- induces apoptotic cell death in primary leukemic cancer cells (Figure 2).
- is a metabolically stable, highly permeable and highly soluble compound. It does not inhibit or induce cytochrome P450 enzymes and is not a P-gp substrate (Table 3).
- has excellent pharmacokinetic properties in preclinical models (Table 3).
- induces a dose-dependent inhibition of tumor growth in various tumor models (Table 4).
- shows highest efficacy in models of hematological malignancies (AML and lymphoma; Figures 3 and 4).

CONCLUSION

These data demonstrate that SB939 is a potent and effective anti-tumor drug in *in vitro* and *in vivo* models of hematological malignancies. It has superior physicochemical, metabolic, pharmacokinetic and pharmacodynamic properties compared to reference HDAC inhibitors that are currently in clinical trials. Pre-clinical PK/PD modeling predicts that SB939 will have superior activity in patients.

SB939 is currently being evaluated in phase I clinical trials as a single agent in both hematological malignancies and solid tumors.

REFERENCES

[1]K. Sangthongpitag, H. Wang, P. Yeo, et.al. (2006) EORTC-NCI-AACR Symposium on Molecular Targets and Cancer Therapeutics Prague Congress Centre, Prague, Czech Republic, Abstract number 166

[2]K. Sangthongpitag, H. Wang, P. Yeo, et.al. (2006) EORTC-NCI-AACR Symposium on Molecular Targets and Cancer Therapeutics Prague Congress Centre, Prague, Czech Republic, Abstract number 162