SB939: a potent and orally active HDAC inhibitor for the treatment of colorectal cancer

K. Sangthongpitag, G. Greicius*, W. Stunkel, K.P. Koh, Y. C. Tan, V.M. Nayagam, Z. Bonday, M. K. Goh, C. P. Ng, K.C. Goh, X.K. Wang, K.L. Goh, W.C. Ong, X.F Wu, C.Y. Hu, H. H. Khng, S. K. Goh, T. Ng, M. Entzeroth, H.S. Wang, N.F Yu, K. Lee, D.Z. Chen, P.L. Lye, J. Chang, M. Ng, W.P. Deng, M. L. Khoo, T. Lu, C.K. Soh, E.T. Sun, P. Yeo, X. Liu, E. Goh, L.S. New, E. Kantharaj, S. Pettersson* and J. Wood

S*BIO Pte. Ltd., 1 Science Park Road #05-09 The Capricorn, Singapore Science Park II, Singapore 117528 WWW.SBIO.COM; *MTC, Division of Molecular Pathology, Laboratory of Gastrobiology, Karolinska Institute, Theorells väg 3171 77 Stockholm

INTRODUCTION

- Colorectal cancer is the second leading cause of cancer-related death in developed countries.
- The etiology of colorectal cancer could arise from chronic inflammation or genetic mutation of polyps, (e.g. *APC*).
- Histone deacetylases (HDAC) regulate a number of genes involved in inflammation, cell proliferation and differentiation, cell cycle progression and cell survival. High expression of HDACs has been demonstrated in human colon cancer[1].
- There are a number of HDAC inhibitors, e.g. SAHA, MS275 and PXD101, emerging as a new class of anticancer agents which have shown good efficacy in clinical trials for treatment of hematological malignancies.
- None of these HDAC inhibitors have demonstrated good anti-tumor activity in solid tumors, which may be due to the their poor pharmacokinetic properties and dose limiting toxicities.
- Here, we present the pharmacological profiling of SB939, a novel compound with improved metabolic, pharmacokinetic and pharmaceutical properties[2], in colon cancer models.

METHODS AND RESULTS

Figure 1: HDACs expression profile and gene knock down using siRNA

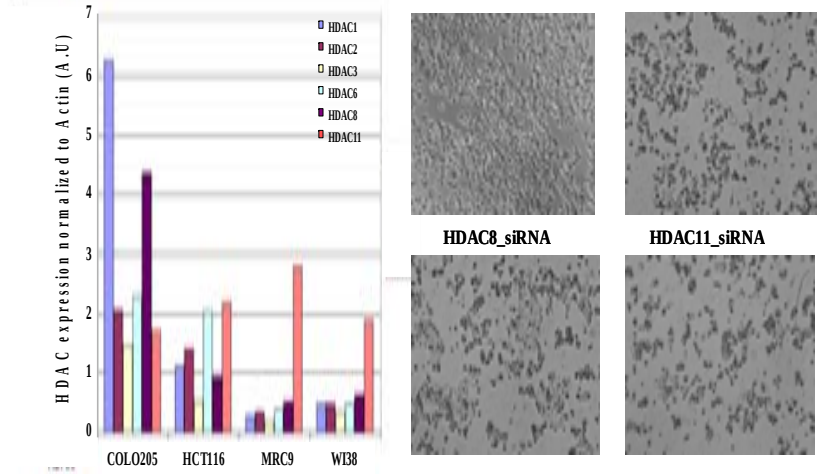
HDAC expression normalized to Actin (A.U)
HDAC1
HDAC2
HDAC3
HDAC6
HDAC8
HDAC11
COLO205
HCT116
MRC9
WI38
Control
HDAC1_siRNA
HDAC8_siRNA
HDAC11_siRNA

- Expression of different HDAC isoforms was quantified using quantitative real-time PCR.
- HDAC1, 2, 3, 6 and 8 were over-expressed in colon cancer cell lines.
- Knockdown of HDAC1, 8, and 11 in a colorectal cancer cell line (HCT116) resulted in changes in cell morphology and an increase in cell death.

Figure 2: Inhibition of recombinant HDAC enzyme activities

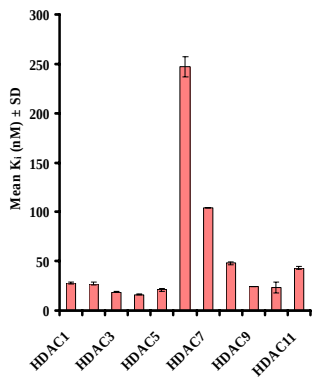
Mean Ki (nM) ± SD
HDAC1
HDAC3
HDAC5
HDAC7
HDAC9
HDAC11

- HDAC enzymatic activities were measured using the Biomol HDAC fluorescent assay.
- SB939 potently inhibited a broad range of class I and class II HDACs with K_i values ranging from 16 to 247 nM.
- SB939 was least potent against HDAC6 and HDAC7 (K_i values of 247 nM and 104 nM respectively).
- SB939 did not inhibit SIRT1, a representative class III HDAC.

Figure 3: Cellular response to SB939 treatment

Colorectal cell lines	IC_{50} (µM; Mean± SD) for inhibition of cell proliferation
Colo205	0.6 ± 0.1 (n = 41)
HCT116	0.5 ± 0.2 (n = 5)
SW620	0.5, 0.5 (n = 2)

- SB939 inhibited cell proliferation of 3 different colorectal cancer cell lines with similar potency.

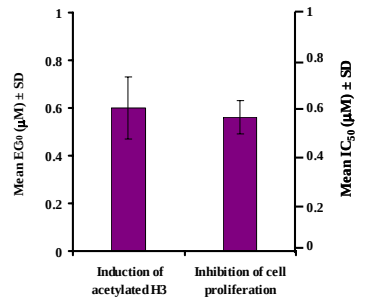
Mean EC50 (µM) ± SD
Mean IC50 (µM) ± SD
Induction of acetylated H3
Inhibition of cell proliferation

- Acetylation of histone3 was detected using an ELISA after 24 h of treatment of Colo205 cells with SB939.
- Induction of acetylation of histone3, (EC_{50} 0.6 ± 0.1 µM) occurred in the same concentration range as inhibition of cell proliferation (IC_{50} 0.6 ± 0.1 µM). Data mean ± SD.

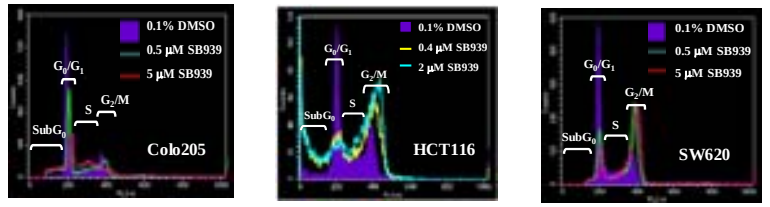
0.1% DMSO
0.5 µM SB939
5 µM SB939
Colo205
0.4 µM SB939
2 µM SB939
HCT116
SW620
SubG0
S
G0/G1
G2/M

- After 24 h of treatment with SB939, colon cancer cells were stained with propidium iodide and characterized using flow cytometry.
- SB939 arrested Colo205 cells at G_1/S phase, HCT116 and SW620 cells at G_2/M phase, and increased apoptotic cells at subG_0 phase.

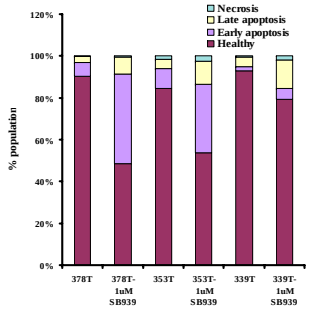
Necrosis
Late apoptosis
Early apoptosis
Healthy
% population
378T
378T-1uM SB939
353T
353T-1uM SB939
339T
339T-1uM SB939

- After 96 h of treatment with SB939, the primary colon cancer cells were stained with Annexin V-FITC and propidium iodide and then analysed by flow cytometry. Cell death was analyzed and quantified using cell-quest software.
- SB939 induced apoptotic cell death in all 3 primary colorectal cancer cell lines.

Figure 4: Profiling of mechanistic biomarkers

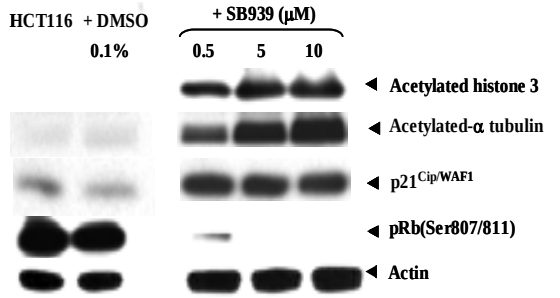
HCT116 + DMSO 0.1%
+ SB939 (µM)
0.5 5 10
Acetylated histone 3
Acetylated-α tubulin
p21Cip/WAF1
pRb(Ser807/811)
Actin

- The molecular effects of SB939 were evaluated in HCT116 cell line by western blot analysis.
- SB939 induced acetylation of histone3 and alpha tubulin, increased expression of the pro-apoptotic protein, $p21^{Cip/WAF1}$, and reduced phosphorylation of retinoblastoma protein.

Figure 5: Induction of acetylation of histone3 in HCT116 tumor tissues

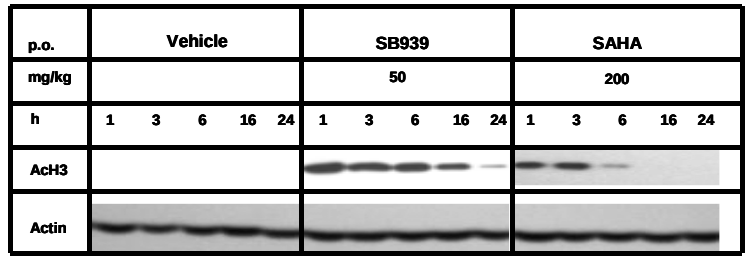
p.o.
Vehicle
SB939
SAHA
mg/kg
50
200
h
1 3 6 16 24
AcH3
Actin

- Acetylation of histone3 in tumor samples was measured by western blot. Each blot represents tumors from 3 mice (10 µg protein each).
- After a single oral dose of SB939 (50 mg/kg) or SAHA (200 mg/kg), acetylation of histone3 was induced in tumor tissue within 1 h after dosing.
- The response persisted longer for SB939 than for SAHA.

Figure 6: Anti-tumor activity of SB939 in a model of advanced human colorectal cancer (HCT116 bearing nude mice)

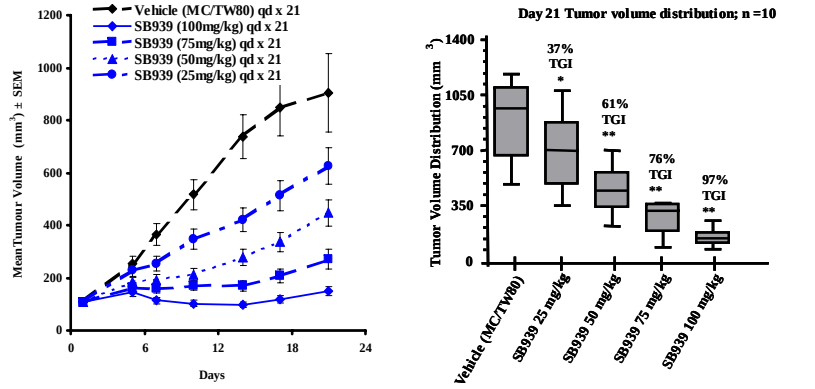
Vehicle (MC/TW80) qd x 21
SB939 (100mg/kg) qd x 21
SB939 (75mg/kg) qd x 21
SB939 (50mg/kg) qd x 21
SB939 (25mg/kg) qd x 21
Mean Tumour Volume (mm3) ± SEM
Days
Day 21 Tumor volume distribution; n =10
Tumor Volume Distribution (mm3)
37% TGI *
61% TGI **
76% TGI **
97% TGI **
Vehicle (MC/TW80)
SB939 25 mg/kg
SB939 50 mg/kg
SB939 75 mg/kg
SB939 100 mg/kg

- After daily oral administration for 21 days at 25, 50, 75 and 100 mg/kg, SB939 induced significant and dose dependent tumor growth inhibition (TGI). Data mean ± SEM.
- All mice tolerated SB939 well. The mean group body weight loss on day 21 was 6%, 8%, 6% and 16% for the 25, 50, 75 and 100 mg/kg doses respectively.

Figure 7: Blood vessel formation in HCT116 tumor tissue

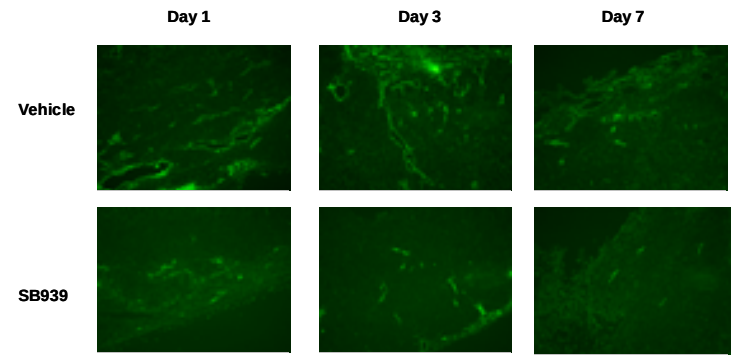
Day 1
Day 3
Day 7
Vehicle
SB939

- SB939 was administered orally to HCT116 tumor bearing mice at a dose of 50 mg/kg for 7 days.
- Lectin was injected into the tail vein of HCT116 tumor bearing mice at different time points. The HCT116 tumor tissue was collected 30 min later.
- SB939 reduced the positive staining of lectin perfused blood vessels in HCT116 tumors.

Figure 8: Anti-tumor activity of SB939 in a mouse spontaneous colon cancer model, *Apc*$^{min/+}$ Mice

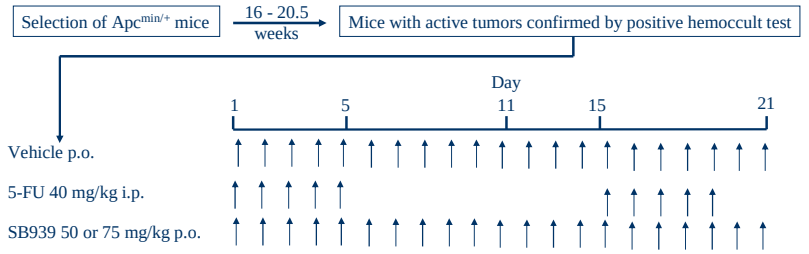
Selection of Apcmin/+ mice
16 - 20.5 weeks
Mice with active tumors confirmed by positive hemoccult test
Day
1
5
11
15
21
Vehicle p.o.
5-FU 40 mg/kg i.p.
SB939 50 or 75 mg/kg p.o.

- SB939 was given orally once daily in doses of 50 or 75 mg/kg to *Apc*$^{min/+}$ mice that were 16 to 20 weeks old and positive in a hemocult test.
- 5-FU was administered at 40 mg/kg daily by intraperitoneal injection for 5 consecutive days followed by 9 days of rest for two cycles.

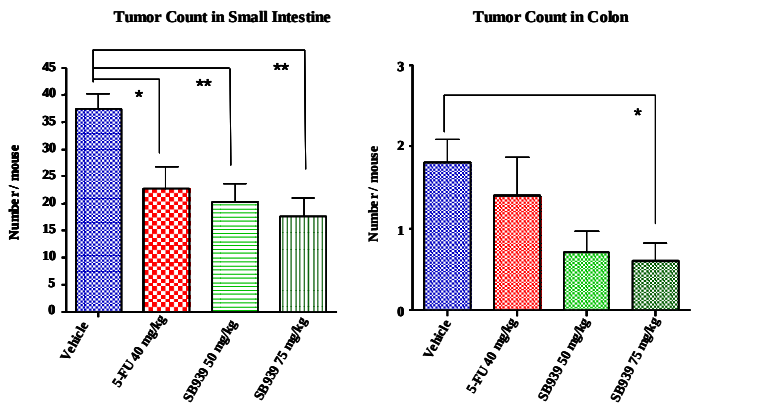
Tumor Count in Small Intestine
Tumor Count in Colon
Number / mouse
Vehicle
5-FU 40 mg/kg
SB939 50 mg/kg
SB939 75 mg/kg

* P < 0.05; ** P < 0.01; n = 10; mean ± SD

- SB939 significantly reduced tumor size and tumor number in both the small intestine and colon (46% and 61% at 50 mg/kg SB939; 53% and 67% at 75 mg/kg SB939 respectively) and was well tolerated.
- 5-FU significantly reduced tumor size and number in the small intestine by 39% but had no significant effect on lesions in the colon.

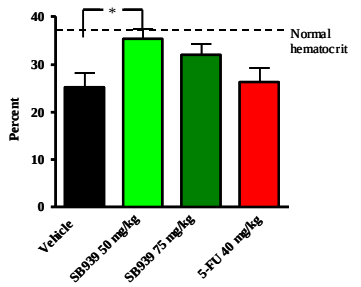
Percent
Normal hematocrit
Vehicle
SB939 50 mg/kg
SB939 75 mg/kg
5-FU 40 mg/kg

- SB939 but not 5-FU significantly increased hematocrit and reduced the number of immature erythrocytes (Ter-119^{+}) in the spleen.

SUMMARY

- HDACs are over-expressed in colon cancer cell lines and knockdown of selective HDAC isoforms using siRNA leads to apoptotic cell death (Figure 1).
- SB939 potently inhibits a broad range of class I and class II HDACs but is less potent against HDAC6 and HDAC7 (Figure 2).
- SB939 inhibits cellular HDAC enzyme activity, resulting in induction of acetylation of histone3 and inhibition of proliferation in colon cancer cell lines (Figure 3).
- SB939, in a dose dependent manner, arrests Colo205 at G_0/G_1 phase, HCT116 and SW620 at G_2/M phase, and increases apoptotic cell death at subG_0 phase (Figure 3).
- SB939 induces acetylation of histone3 and alpha tubulin, increases expression of the cyclin dependent kinase inhibitor and pro-apoptotic protein, $p21^{Cip/WAF1}$ in HCT116 colon cancer cells (Figure 4).
- SB939 induces H3 acetylation in HCT116 tumor tissues at 1 h after administration of an acute oral dose (50 mg/kg). The duration of this response persists for up to 24 h and is longer than that induced by SAHA at 200 mg/kg (Figure 5).
- SB939 induces a dose-dependent inhibition of tumor growth in a model of advanced human colorectal cancer (HCT116) (Figure 6).
- SB939 reduces the formation of blood vessels in HCT116 tumors (Figure 7).
- SB939 reduces tumor size and tumor count, and reduces progression of anemia in *Apc*$^{min/+}$ mice, a model of early stage colorectal cancer (Figure 8).

CONCLUSION

SB939 is an effective anti-tumor drug in both *in vitro* and *in vivo* colon cancer models and has the potential to be an effective oral therapy for early and late stage colorectal cancer. SB939 is currently being evaluated in phase 1 clinical trials .

REFERENCES

[1]B.H. Huang, M. Laban, C Leung et.al. (2005) Cell Death Differ 12; 395-404

[2]K. Sangthongpitag, H.S Wang, P. Yeo, et.al. (2006) EORTC-NCI-AACR Symposium on Molecular Targets and Cancer Therapeutics Prague Congress Centre, Prague, Czech Republic, Abstract number 166