Abstract # 2924

AACR: 100th Annual Meeting, 2009

Pharmacokinetic/Pharmacodynamic (PK/PD) relationships of novel HDAC inhibitors in an HCT-116 mouse xenograft tumor model

Ramesh Jayaraman, Mohammed Khalid Pasha, Pauline Yeo, Kanda Sangthongpitag, Haishan Wang, Hannes Hentze, Veronica Novotny-Diermayr, Jeanette Wood and Kantharaj Ethirajulu

*S*BIO Pte Ltd, 1 Science Park Road, # 05-09 The Capricorn, Singapore Science Park II, Singapore 117528. www.sbio.com*

INTRODUCTION

Inhibition of Histone deacetylase (HDAC) activity leads to cell cycle arrest and terminal differentiation of cancer cells, and represents a promising approach in the treatment of cancer[1].

- Clinical proof-of-concept was shown with the first HDAC inhibitor Zolinza (SAHA), now approved for cutaneous T-cell lymphoma (CTCL). Other HDAC inhibitors like FK228, PXD101, LBH589 and MGCD0103 are in clinical trials.
- Pre-clinical PK/PD relationships are not well characterized for inhibitors of HDACs. PK/PD relationships are critical for setting dose and regimen in pre-clinical and clinical stages of drug discovery[2].
- The aim of this study was to explore relationships between PK and PD parameters, based on established paradigms used in anti-infective drug discovery
- PK parameters (AUC, C_{max} and $t_{1/2}$) were correlated with PD parameters (tumor cell proliferation inhibition [$IC_{50,HCT116}$] and tumor growth inhibition [TGI]) in a human colorectal cancer (HCT-116) xenograft model in nude mice treated with a series of novel HDAC inhibitors and reference compounds.

METHODOLOGY

Compounds: SB939,SB869, SB207, LBH589, SAHA, PXD101

Inhibition of HDAC1 activity ($IC_{50,HDAC1}$): IC_{50} values were determined against HDAC1 (in triplicates) using an *in vitro* fluorescence-based HDAC assay in 96-well format (BIOMOL Inc, Boston, USA) **(Table 1)**

***In vitro* cell proliferation assay ($IC_{50,\ HCT\text{-}116}$):** HCT-116 cells (ATCC, Virginia, U.S.A.) were seeded in 96-well plates at 3000 cells/well and then incubated at 37°C, 5% CO_2 for 24 h. Cells were treated with compounds at various concentrations for 96 h and cell proliferation was monitored using the CyQUANT™ assay (Invitrogen Pte Ltd). IC_{50} values were estimated from dose response curves (XL-fit, IDBS, New Jersey, U.S.A) **(Table 1)**

Pharmacokinetics: *Animals:* Female Balb/c nude mice, 18-22 gm, 8-10 weeks. ***Dose:*** 50 mg/kg. ***Route:*** Oral (gavage). ***Formulation:*** 0.5% methyl cellulose and 0.1% Tween-80 in water. ***Time points:*** Blood samples were taken at pre-dose, 5 , 10, 30 min, 1 h, 2, 4, 8, and 24 h by cardiac puncture following CO_2 anesthesia. ***Processing and bioanalysis:*** Blood samples were centrifuged for 10 min at 3000 rpm and plasma harvested and analyzed by LC-MS/MS[3] . ***PK Parameters:*** Non-compartmental Analysis, WinNonlin (ver. 5.1, Pharsight) **(Table 1).** PK parameters for 100 mg/kg SB939 and PXD101, and 200 mg/kg SAHA were estimated by extrapolation of PK data from 50 mg/kg, assuming dose proportionality.

Table 1: Summary of PK/PD parameters

PK/PD Attributes	SB939	SAHA	PXD101	LBH589	SB207	SB869
$IC_{50,\ HDAC\ 1}$ (ng/ml)	25	32	19	2.4	15	18
$IC_{50,\ HCT\text{-}116}$ (ng/ml)	190	875	185	22	1372	312
Dose (mg/kg)	100	200	100	50	100	50
AUC (ng*h/ml)	3682	2476	514	126	6118	1875
C_{max} (ng/ml)	5264	2004	987	116	6565	3980
$AUC/IC_{50,\ HCT\text{-}116}$	19	3	3	6	5	6
$C_{max}/IC_{50,\ HCT\text{-}116}$	28	2	5	5	5	13
$T>IC_{50,\ HCT\text{-}116}$ (min)	240	60	30	60	60	90
TGI (%)	97	40	35	50	57	68

In vivo* Xenograft study: *Animals: Female Balb/c nude mice, 10-12 weeks. ***Cell line:*** Mice were implanted *s.c.* with 5 x 10^6 HCT-116 cells. Treatment began when tumor volumes were 75-144 mm^3. ***Doses:*** SB939 (50 and 100 mg/kg), LBH589 (50 mg/kg), SAHA (200 mg/kg) and PXD101 (50 and 100 mg/kg), SB869 and SB207 at 50 mg/kg. ***Regimen***: *q.d.* for 15 days. ***Route:*** Oral (gavage). ***Formulation:*** 0.5% methyl cellulose and 0.1% Tween-80 in water.

Tumor Growth Inhibition: (%TGI) = $[(C_t\text{-}T_t)/(C_t\text{-}C_{t1})*100]$, where, C_t = median tumor size of vehicle control group at time t, T_t = median tumor size of treatment group at time t, C_{t1} = median tumor size of vehicle control group first day of treatment **(Table 1).**

PK/PD parameters: The ratios AUC/ $IC_{50,\ HCT\text{-}116}$, C_{max}/ $IC_{50,\ HCT\text{-}116}$, T> $IC_{50,\ HCT\text{-}116}$ were estimated **(Table 1)** and plotted individually against % TGI for each drug and the r^2 estimated **(Figures 1, 2 and 3).**

SUMMARY OF KEY FINDINGS

1. T >$IC_{50,HCT\text{-}116}$ (r^2=0.80, Figure 1) and AUC/$IC_{50,HCT\text{-}116}$ (r^2=0.76, Figure 2) showed high correlation with %TGI followed by $C_{max}/IC_{50,HCT\text{-}116}$ (r^2=0.71, Figure 3).
2. Anti-tumor activity (%TGI~ 50) was observed when HDAC inhibitors achieved T>$IC_{50,HCT\text{-}116}$ > 1.5 h, AUC/$IC_{50,HCT\text{-}116}$ > 5 and, $C_{max}/IC_{50,HCT\text{-}116}$ >10.
3. SB939 at 100 mg/kg had the highest anti-tumor efficacy (%TGI=97) and the highest T >$IC_{50,HCT\text{-}116}$ (4 h), AUC/$IC_{50,HCT\text{-}116}$ (19), and $C_{max}/IC_{50,HCT\text{-}116}$ (28) when compared to SAHA, PXD101, and LBH589 **(Table 1).**

CONCLUSIONS

- Our data show that the superior preclinical profile and PK/PD relationships of SB939, compared to other HDAC inhibitors, has been translated in Phase 1 clinical trials[4].
- Preclinical PK/PD studies, as we have described here, can help in rapid selection of the best candidates for advancing into clinical trials.

REFERENCES

1. Saverio, M., and Pier, G. P. Histone deacetylase inhibitors and the promise of epigenetic (and more) treatments of cancer. Nature Rev. Cancer, **2006**, 6 :38-51
2. Derendorf, H., Lesko, L. J., Chaikin, P., Colburn, W. A., Lee, P., Miller, R., Powell, R., Rhodes, G., Stanski, D., and Venitz, J. Pharmacokinetic/Pharmacodynamic modeling in drug research and development, J. Clin. Pharmacol., **2000**, 40: 1399-1418
3. Yeo, P., Xin, L., Goh, E., New, L. S., Zeng, P., Wu, X., Venkatesh, P., and Kantharaj, E. Development and validation of high-performance liquid chromatography-tandem mass spectrometry assay for 6-(3-benzoyl-ureido)-hexanoic acid hydroxyamide, a novel HDAC inhibitor, in mouse plasma for pharmacokinetic studies, Biomedical Chromatography **2007**, 21(2): 184 -189
4. Yong, W-P., Goh, B-C., Ethirajulu, K., Yeo, P., Otheris, O., Chao, S. M., Soo, R., Yeo, W-L., Seah, E., and Zhu, J. SB939 Phase 1 Study: A phase I dose escalation study of oral SB939 when administered thrice weekly (every other day) for 3 weeks in a 4-week cycle in patients with advanced solid malignancies (Abstract No. 413), EORTC conference in Geneva, October 2008**.** European J Cancer, **2008**, 6(12): 130.

Figure 1: %TGI versus $T>IC_{50,\ HCT\text{-}116}$

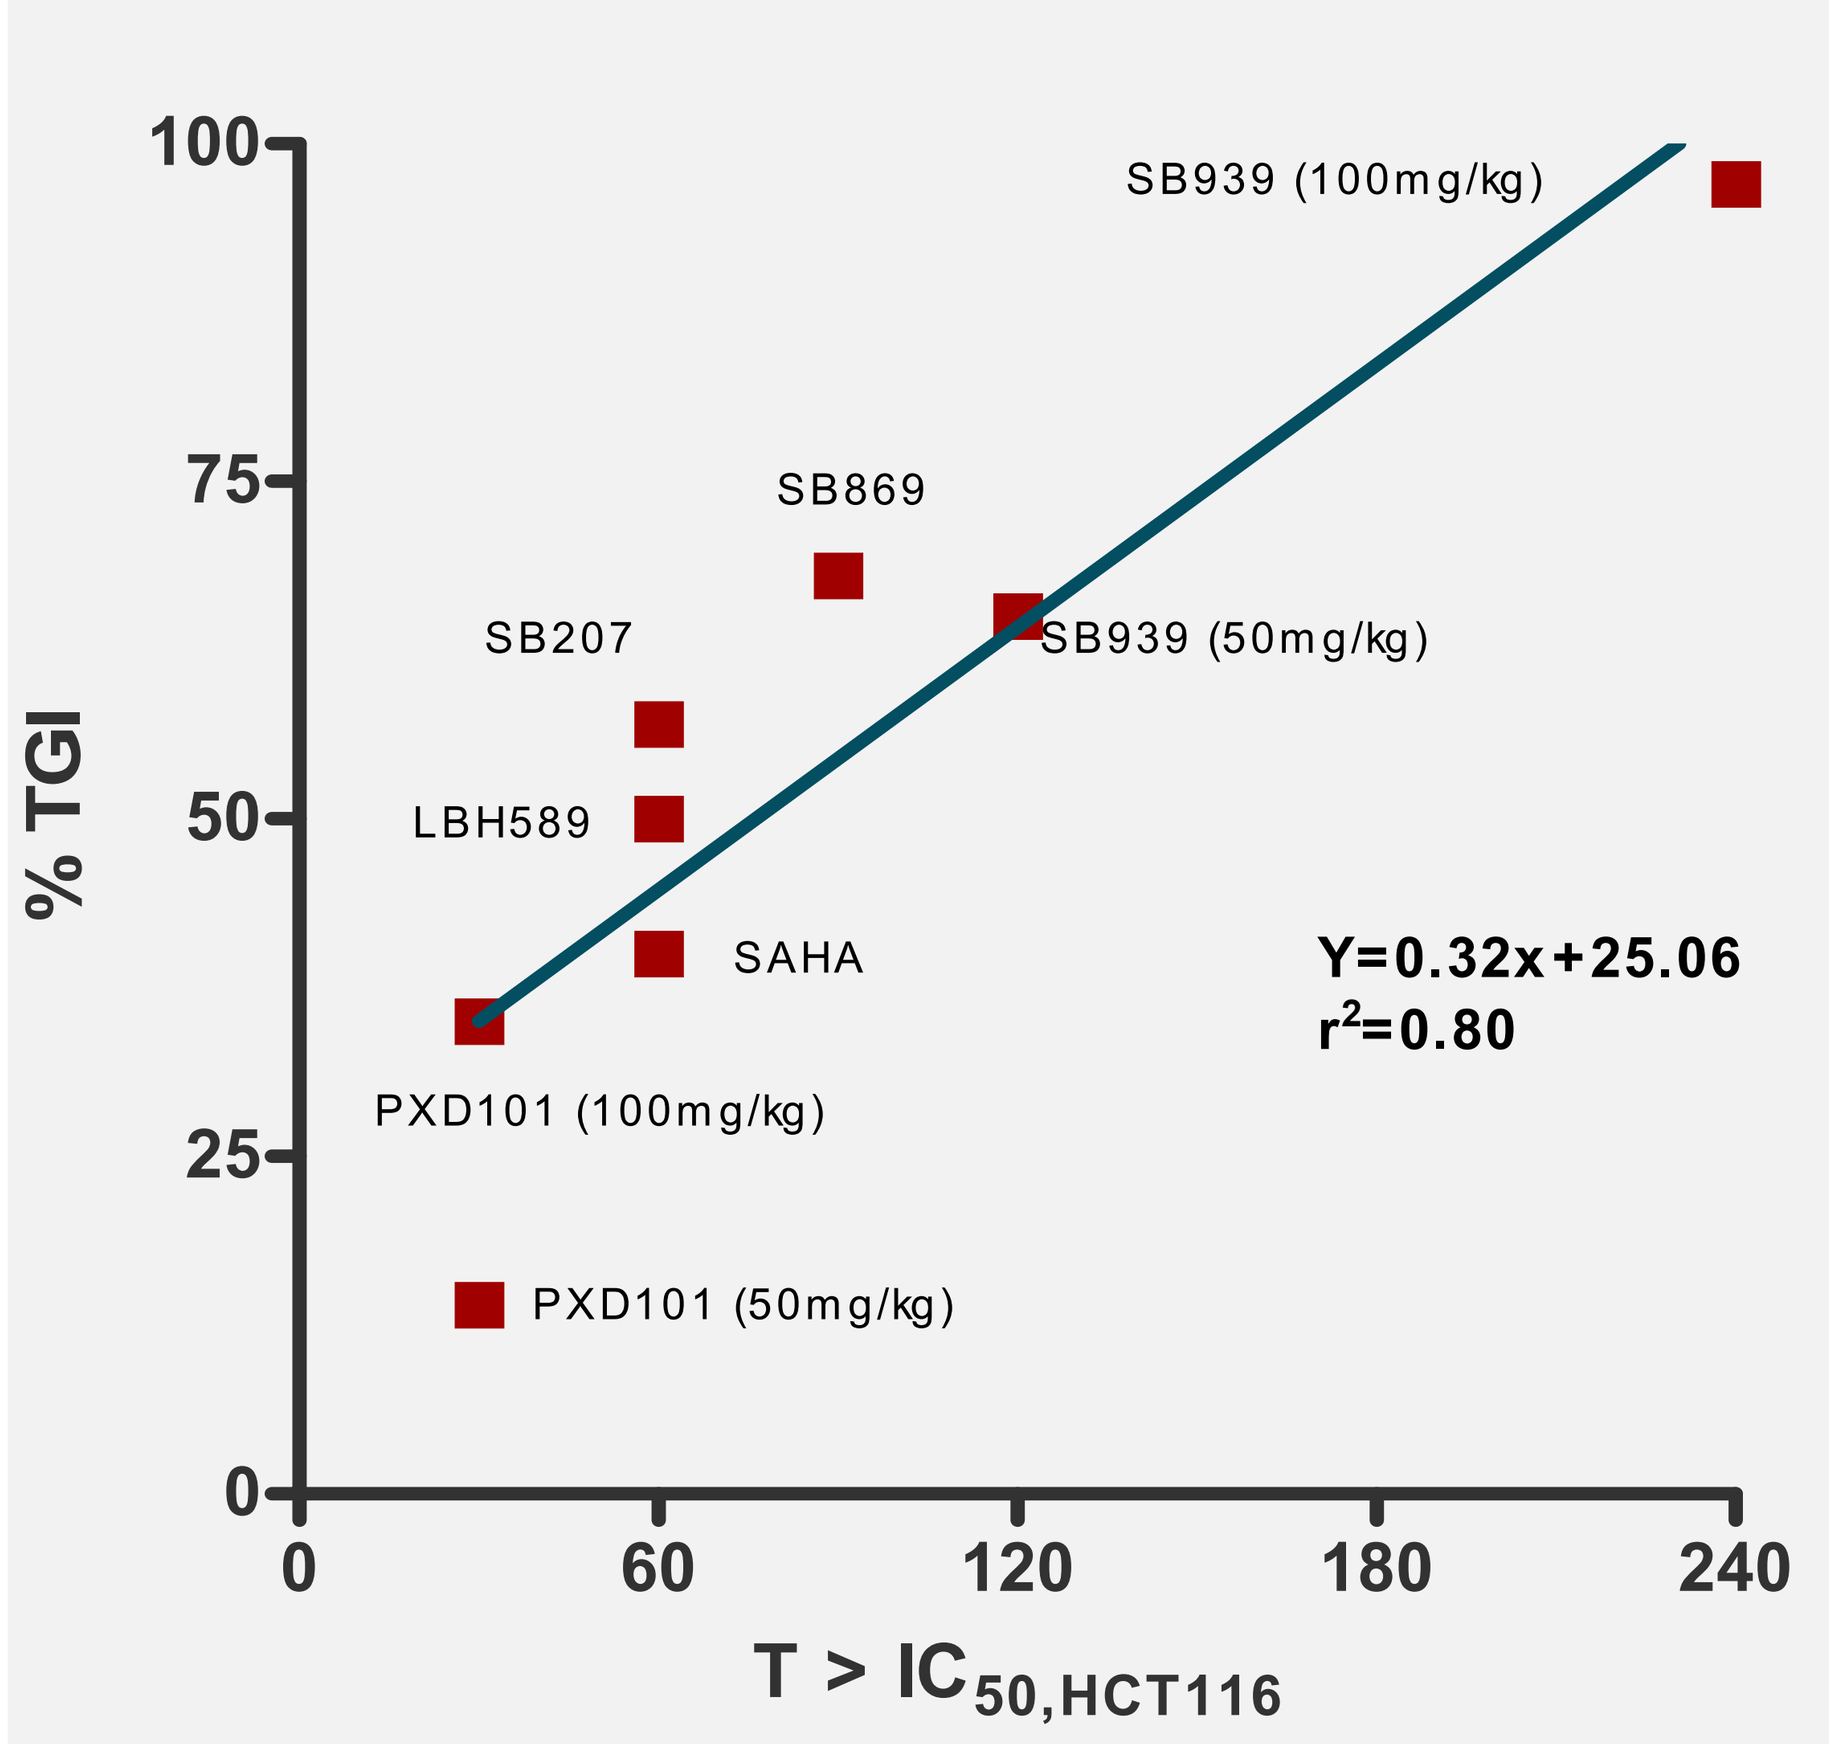
100
75
50
25
0
% TGI
SB939 (100mg/kg)
SB869
SB207
SB939 (50mg/kg)
LBH589
SAHA
PXD101 (100mg/kg)
PXD101 (50mg/kg)
Y=0.32x+25.06
r2=0.80
0
60
120
180
240
T > IC50,HCT116

Figure 2: %TGI versus $AUC/IC_{50,\ HCT\text{-}116}$

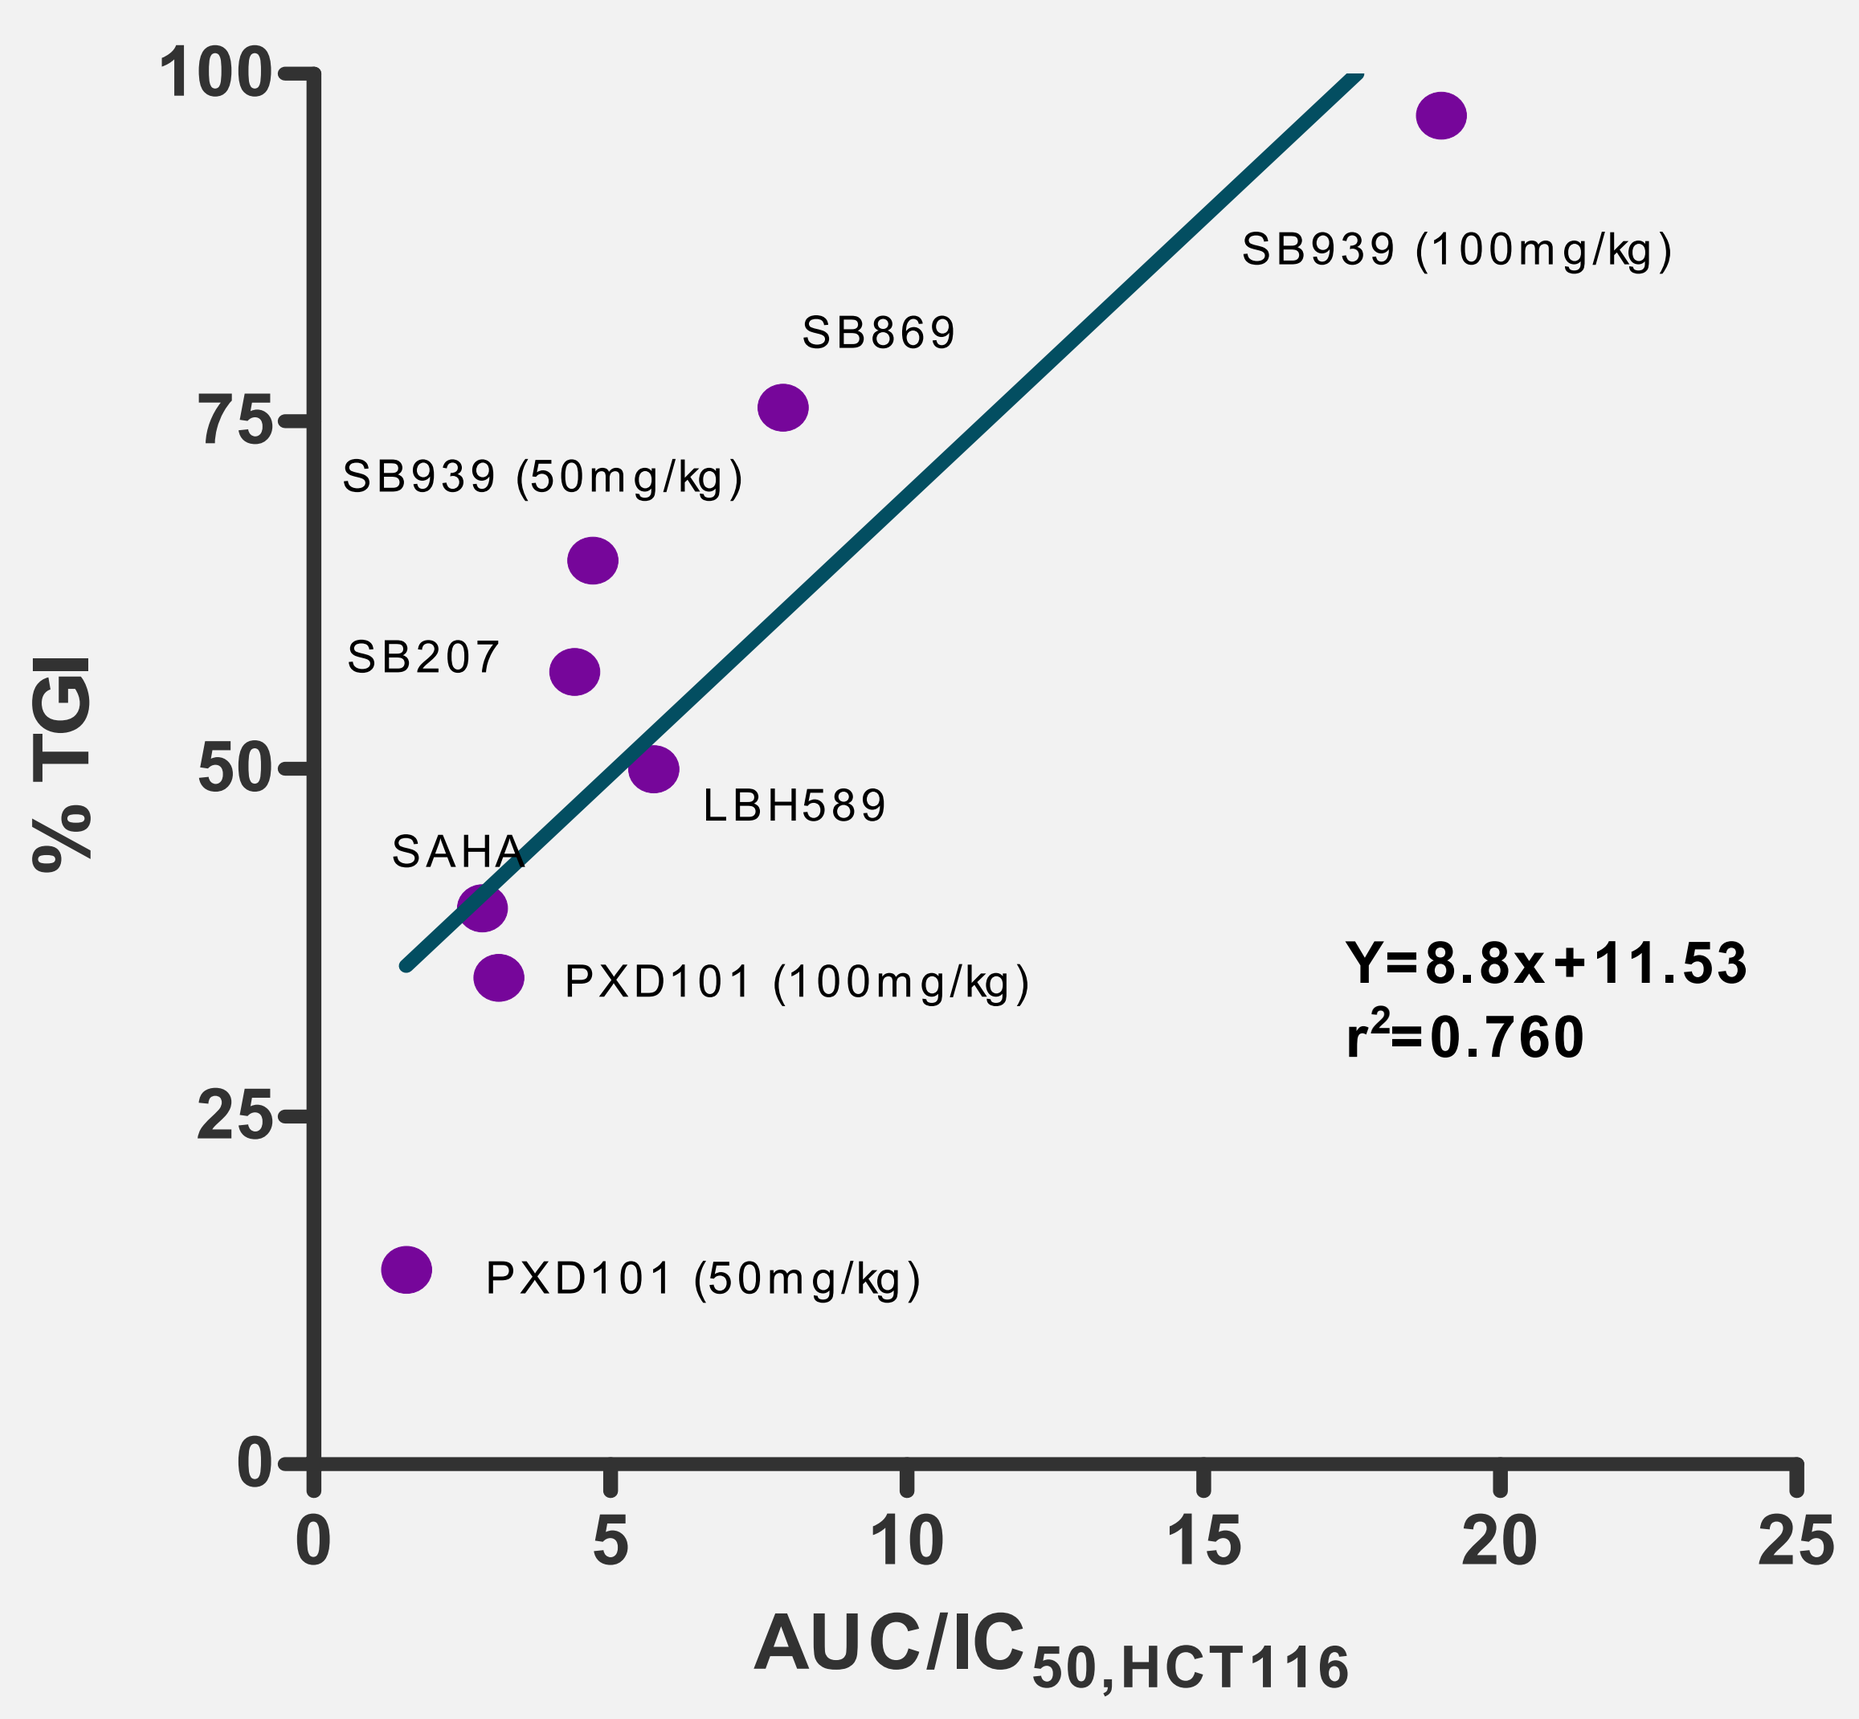
100
75
50
25
0
% TGI
SB939 (100mg/kg)
SB869
SB939 (50mg/kg)
SB207
LBH589
SAHA
PXD101 (100mg/kg)
PXD101 (50mg/kg)
Y=8.8x+11.53
r2=0.760
0
5
10
15
20
25
AUC/IC50,HCT116

Figure 3: %TGI versus $C_{max}/IC_{50,\ HCT\text{-}116}$

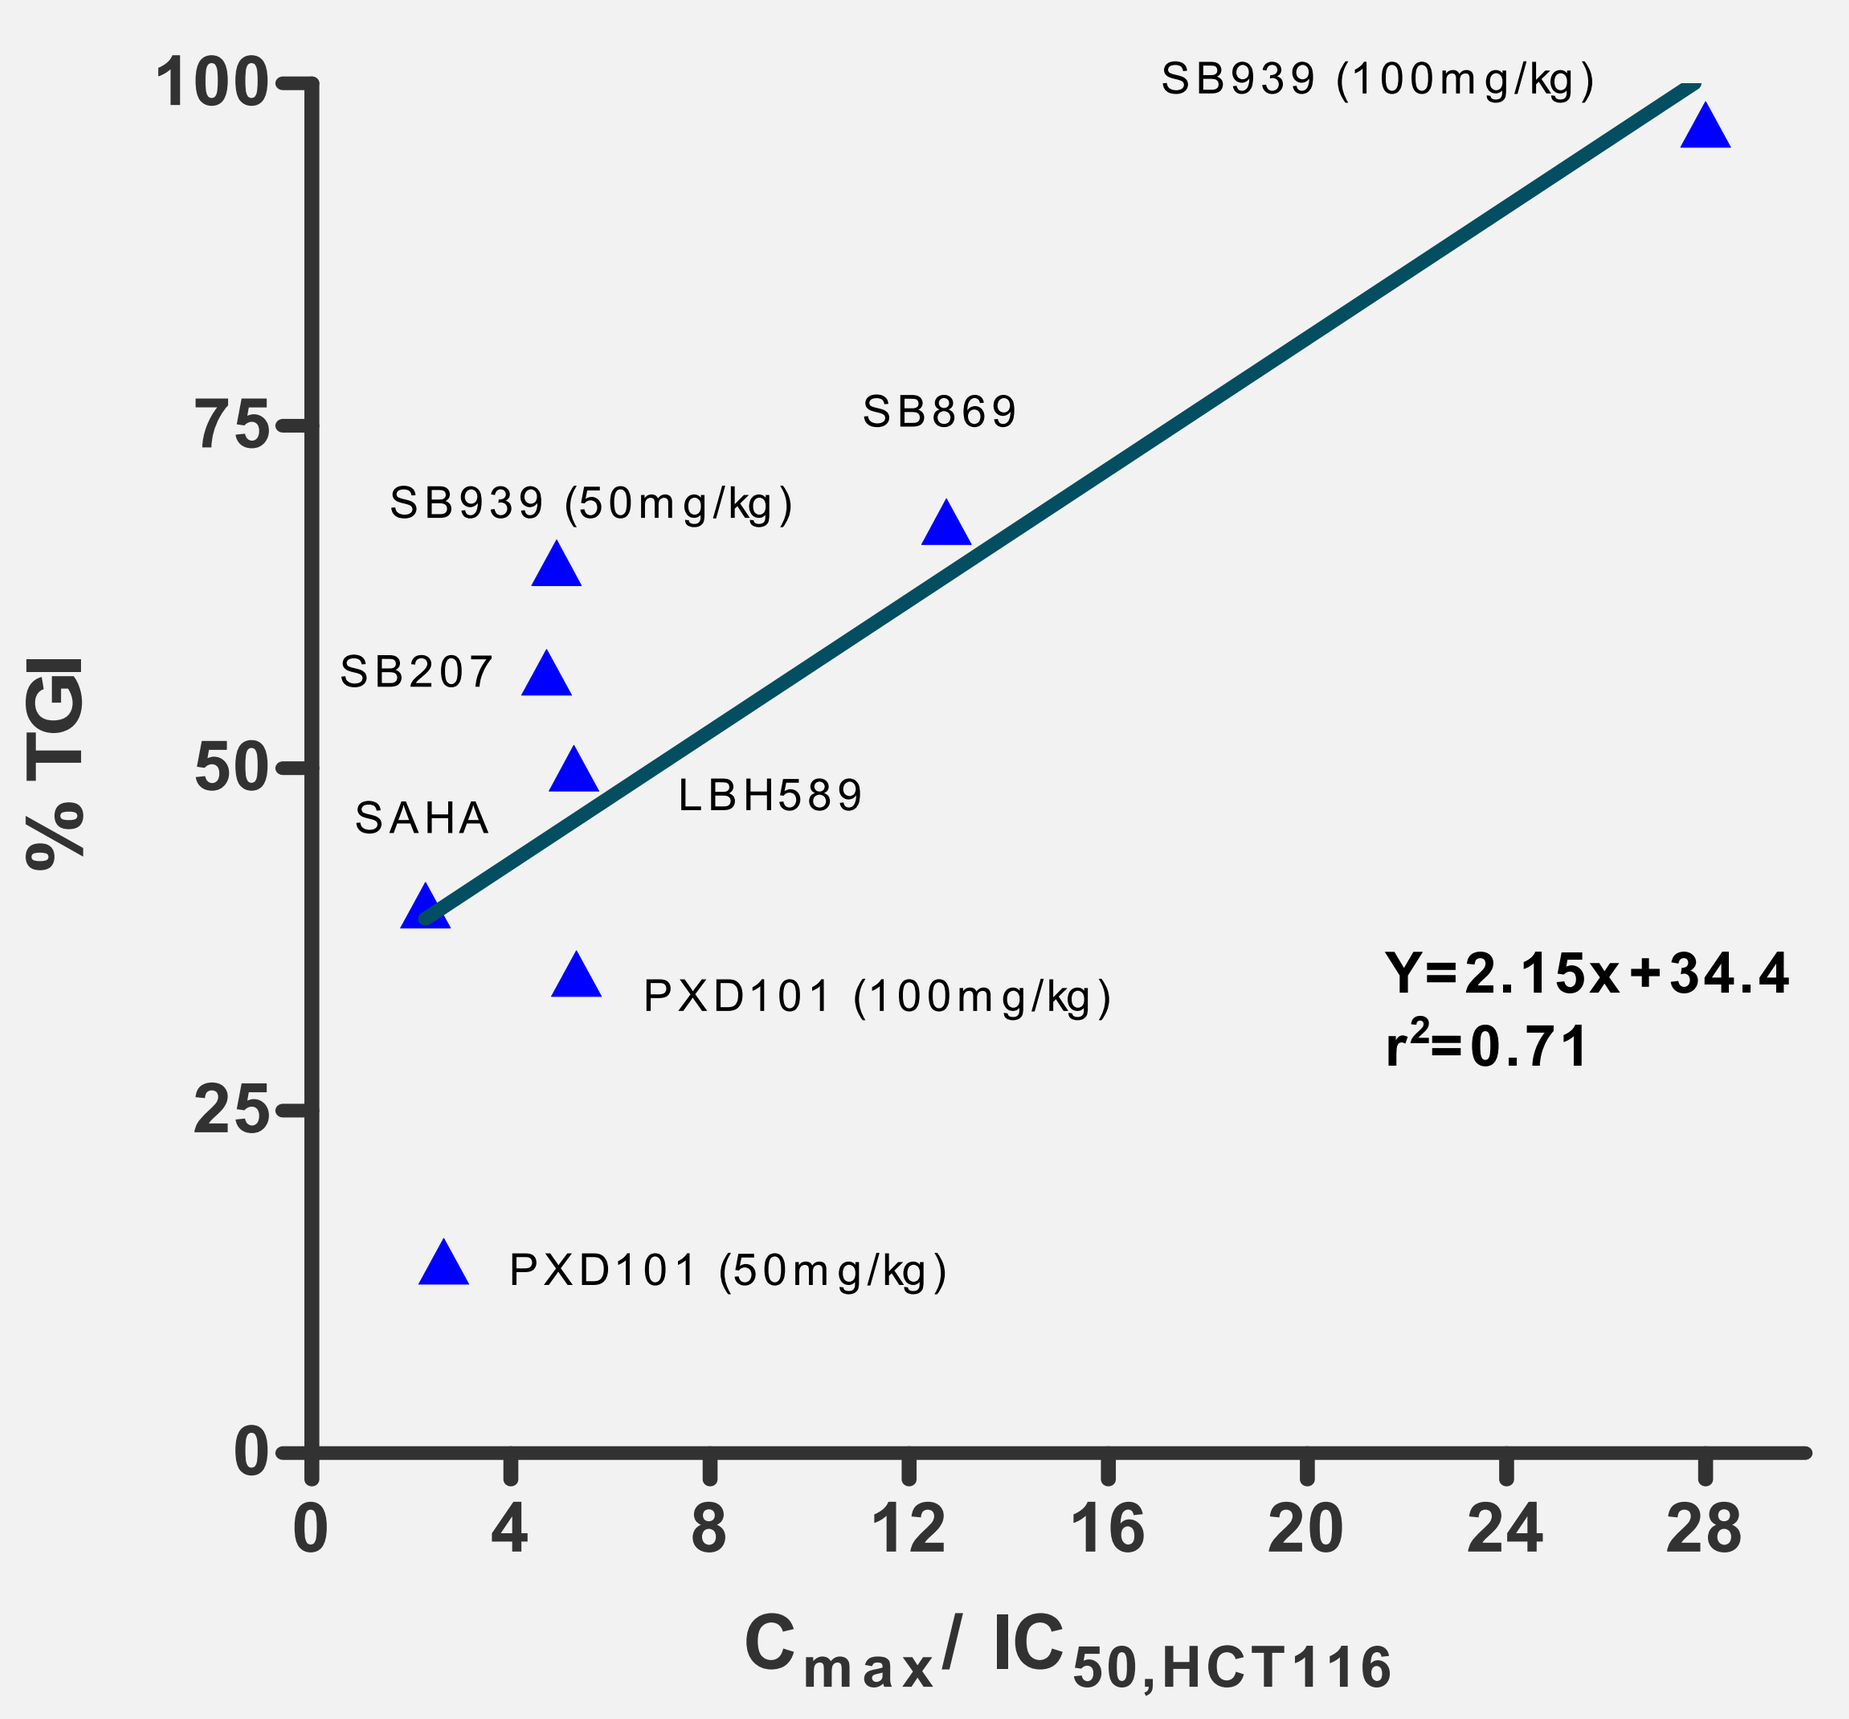
100
75
50
25
0
% TGI
SB939 (100mg/kg)
SB869
SB939 (50mg/kg)
SB207
SAHA
LBH589
PXD101 (100mg/kg)
PXD101 (50mg/kg)
Y=2.15x+34.4
r2=0.71
0
4
8
12
16
20
24
28
Cmax/ IC50,HCT116